Combination of the MCL1 Inhibitor PRT1419 and SMARCA2 Degrader PRT3789 Shows Combinatorial Benefit in SMARCA4-Deleted Lung Cancer

Norman Fultang, Neha Bhagwat, Diane Heiser, Alexander Grego, Michael Hulse, Venkat Thodima, Koichi Ito, Kris Vaddi, Bruce Ruggeri, Peggy Scherle

Prelude Therapeutics Incorporated, Wilmington, DE; contact: nfultang@preludetx.com

Prelude THERAPEUTICS

420

Background

- Induced myeloid leukemia cell differentiation protein (MCL1) is a member of the B-cell lymphoma-2 (BCL2) family of apoptosis regulators and plays a critical role in promoting cancer cell survival.[1]
- PRT1419 is a novel, potent, and orally bioavailable MCL1 inhibitor that demonstrates anti-tumor efficacy in various preclinical models and is currently under evaluation in phase 1 clinical trials in various hematologic and solid malignancies.
- Gene dependency analysis to identify biomarkers of MCL1 inhibitor sensitivity revealed that lung and ovarian cancer cell lines with damaging mutations in SWItch/Sucrose Non-Fermentable (SWI/SNF) ATPase, SWI/SNF Related, Matrix Associated, Actin Dependent Regulator Of Chromatin, Subfamily A, Member 4 (SMARCA4), display strong functional dependency on MCL1.
- The SWI/SNF chromatin remodeling complex functions as a tumor suppressor in a number of cancers. Alterations in subunits of this complex occur in >20% of human cancers, driving tumorigenesis and cancer progression.
- Mammalian SWI/SNF complexes contain one of two catalytic ATPases: SMARCA4 or SMARCA2. Cancers with damaging mutations in SMARCA4 are highly dependent on SMARCA2. We and several others have shown potent synthetic lethality with the use of SMARCA2-targeted protein degraders in SMARCA4-deleted cancers.[2]
- A previously published genome-wide CRISPR screen in SMARCA4-mutant lung cancer demonstrated that loss of MCL1 sensitizes cells to SMARCA2 degradation.[3]

Key Findings

- Here we demonstrate potent synergy between MCL1 inhibitor PRT1419 and a novel, selective, SMARCA2 degrader, PRT3789, in repressing tumor growth in preclinical SMARCA4-deleted lung cancer models.

Results

Figure 1. Damaging Mutations in SMARCA4 and ARID1A Are Associated With MCL1 Dependency in Lung and Ovarian Cancer

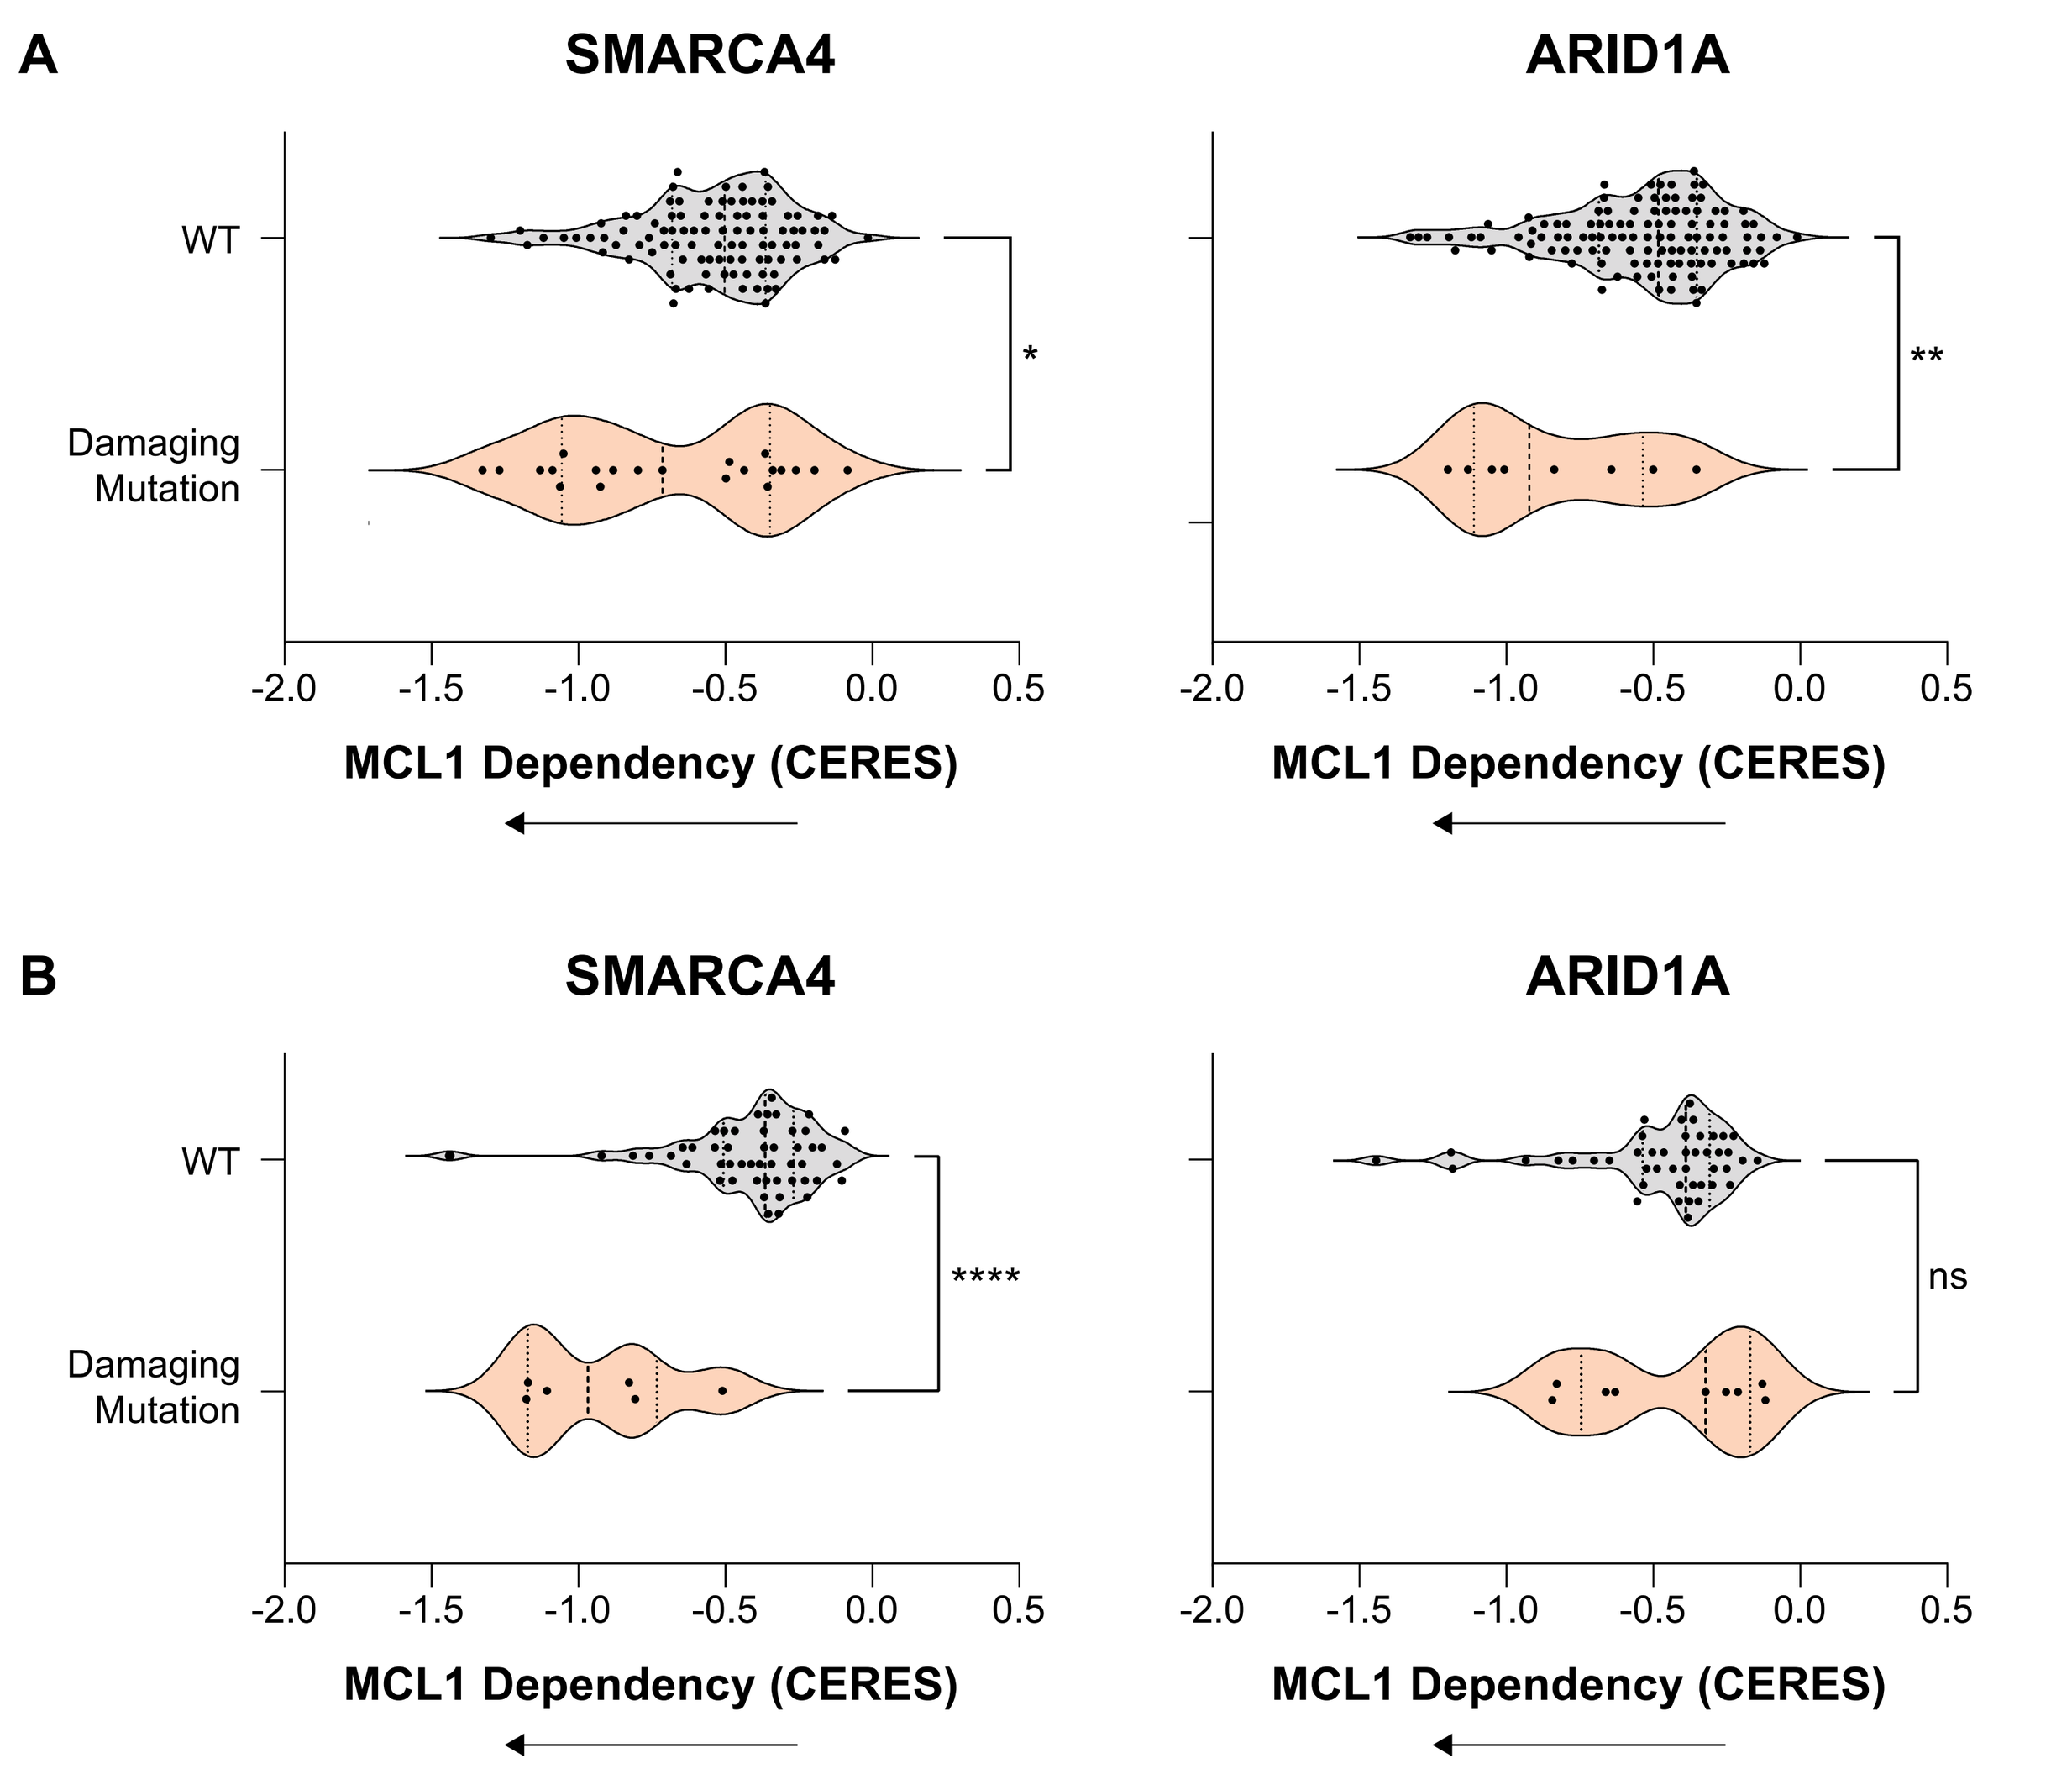
A
SMARCA4
ARID1A
WT
Damaging Mutation
*
**
MCL1 Dependency (CERES)
B
SMARCA4
ARID1A

ns
MCL1 Dependency (CERES)

MCL1 gene dependency analysis in **(A)** lung and **(B)** ovarian cancer cell lines with damaging SMARCA4 and ARID1A mutations. Dependency scores (CERES) were retrieved from DepMap (https://depmap.org). *P<0.05, **P<0.01, ****P<0.0001 versus WT by t-test. ARID1A, AT-Rich Interaction Domain 1A; ns, significant; WT, wild-type.

Figure 2. SMARCA2 Degradation Synergizes With MCL1 Inhibition in SMARCA4-Deleted Lung Cancer *In Vitro*

A

H1568 (SMARCA4-del) — Concentration of PRT3789 (nM) (columns); Concentration of PRT1419 (nM) (rows)

PRT1419 (nM) \ PRT3789 (nM)	0	1.23	3.7	11.11	33.33	100
0	88.0	40.8	33.9	28.9	22.7	25.9
41.2	90.7	44.8	21	19.1	20.2	17.7
123.5	74.2	16.2	7.0	4.8	5.8	5.2
370.4	39.9	6.3	1.7	1.4	2.2	1.8
1111.1	15.3	0.2	0.5	0	0.4	0.5
3333	7.3	0	-0.1	0	0	0.1
10,000	1.8	-0.1	-0.2	-0.2	-0.2	0.3

H520 (SMARCA4-WT) — Concentration of PRT3789 (nM) (columns); Concentration of PRT1419 (nM) (rows)

PRT1419 (nM) \ PRT3789 (nM)	0	1.23	3.7	11.11	33.33	100
0	96.4	97.0	98.6	94.6	96.0	84.7
41.2	97.2	104.1	102.8	102.2	101.2	110.2
123.5	98.3	98.1	99.1	91.2	93.4	87.9
370.4	97.3	84.9	79.1	88.9	90.1	88.6
1111.1	86.4	91.1	74.3	84.5	81.7	84.0
3333	75.1	60.3	65.4	66.3	66.3	53.9
10,000	18.9	19.2	17.0	16.7	18.7	12.6

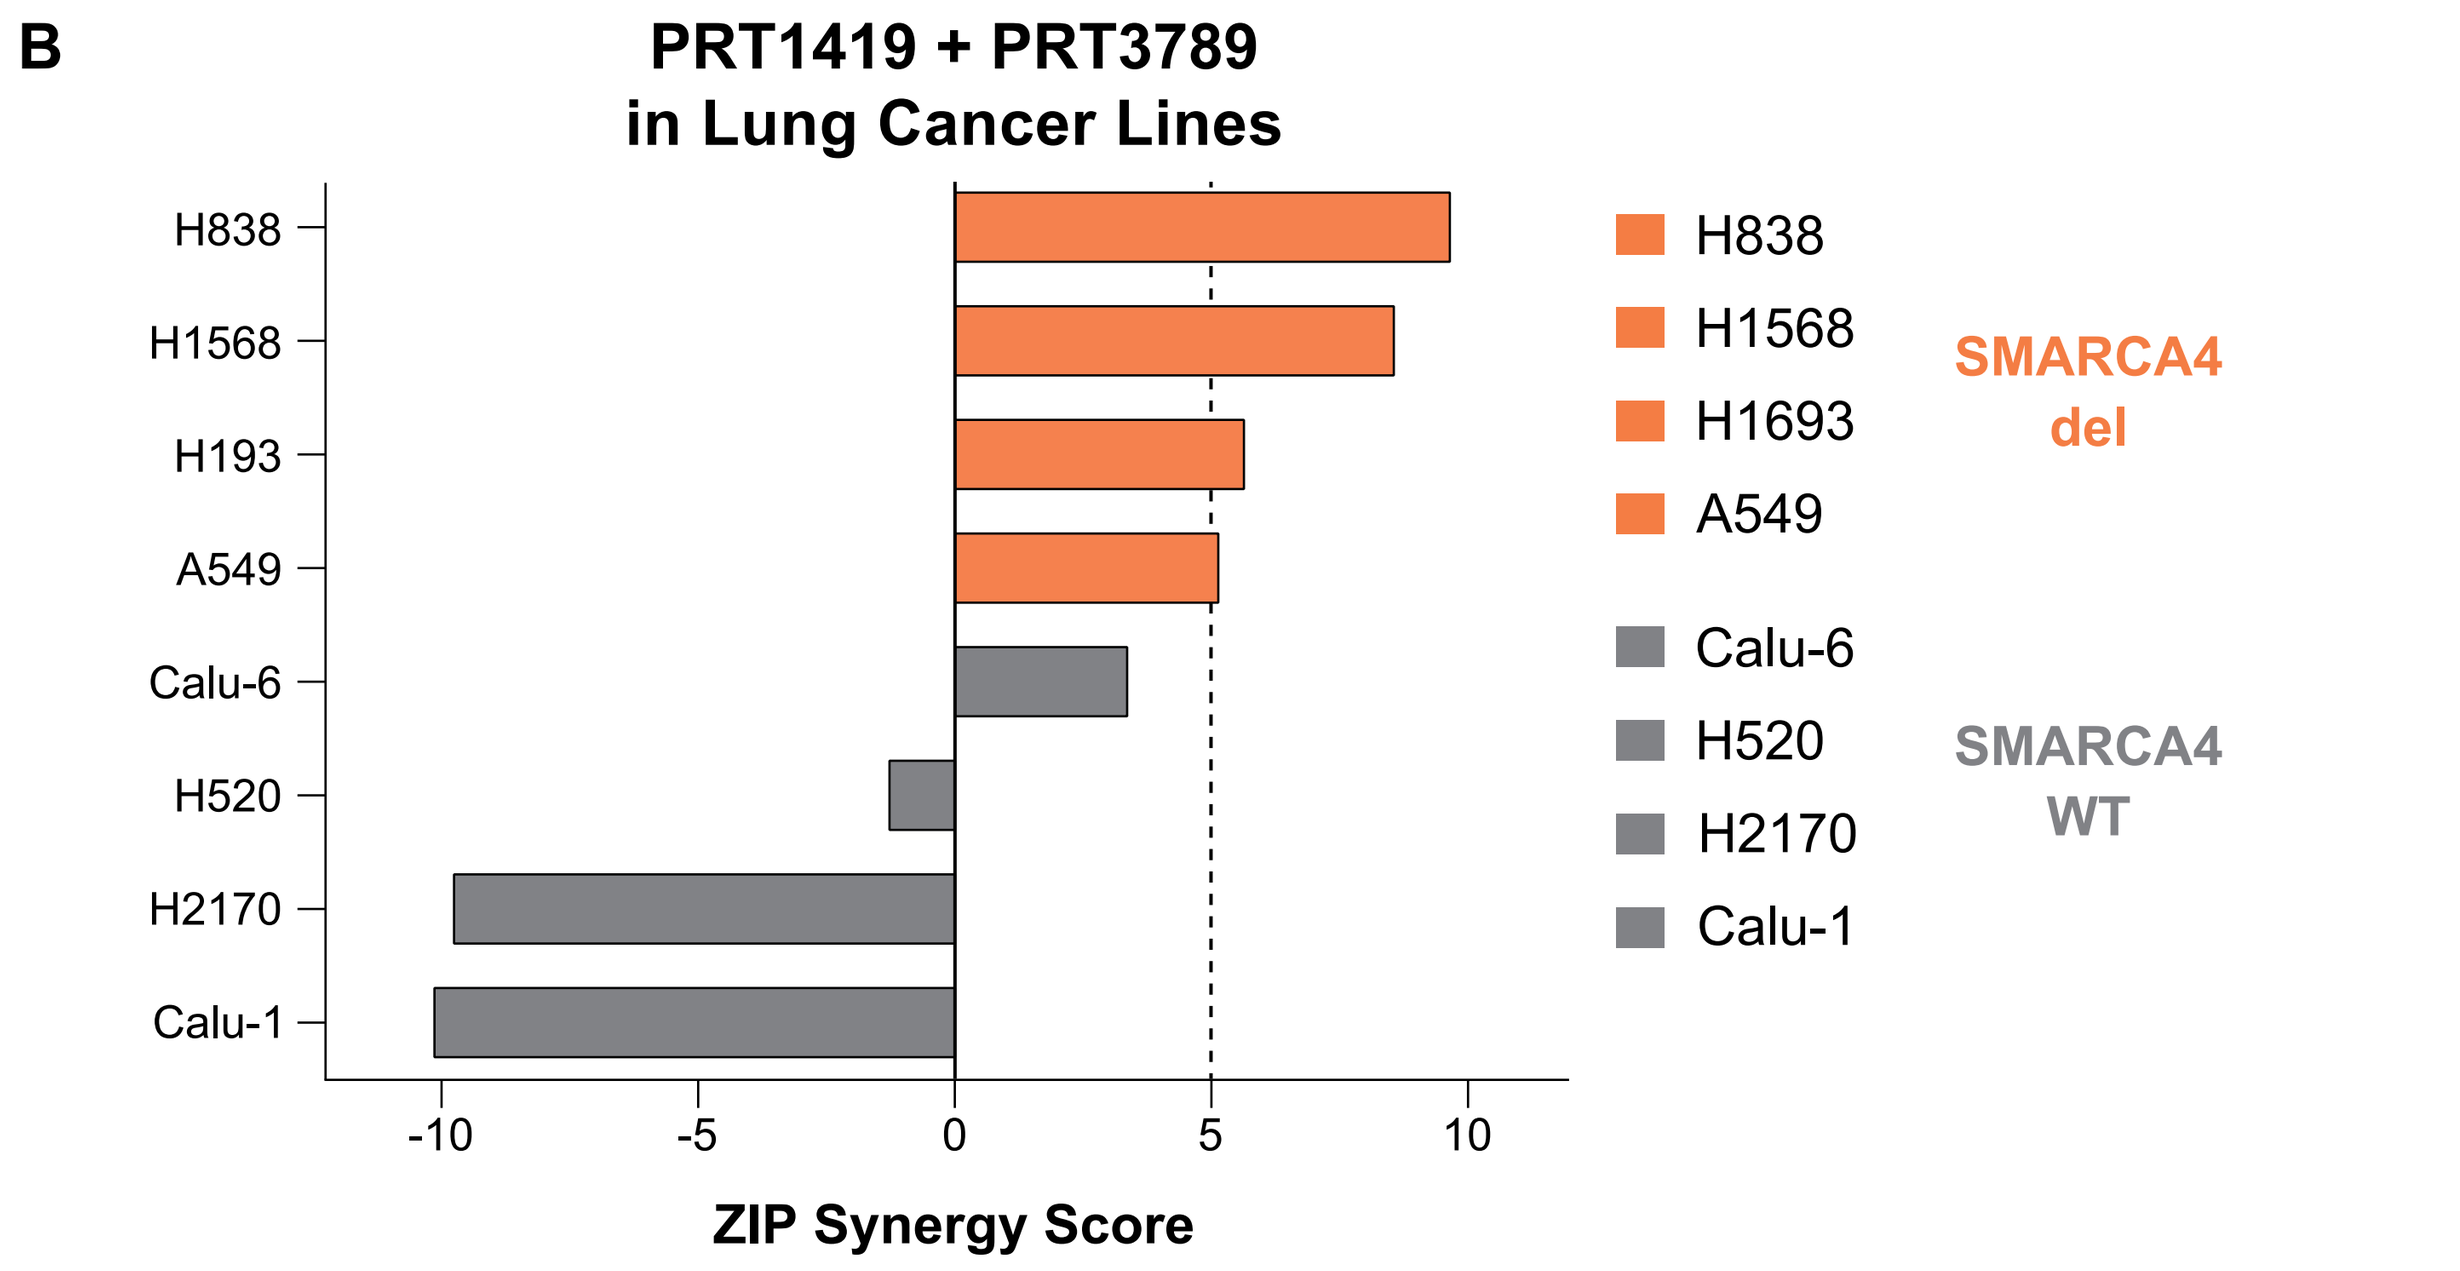
B
PRT1419 + PRT3789
in Lung Cancer Lines
H838
H1568
H193
A549
Calu-6
H520
H2170
Calu-1
ZIP Synergy Score
H838
H1568
H1693
A549
SMARCA4 del
Calu-6
H520
H2170
Calu-1
SMARCA4 WT

(A) CellTiter-Glo assay assessing synergistic inhibition of proliferation in SMARCA4-del and WT cell lines following treatment with SMARCA2 degrader PRT3789 and MCL1 inhibitor PRT1419. Values shown are cell viability calculated as percentage of vehicle (DMSO) control. **(B)** ZIP synergy scores from synergy proliferation assays were computed using SynergyFinder 2.0 (https://synergyfinder.fimm.fi/). Del, deletion; DMSO, dimethyl sulfoxide; ZIP, zero interaction potency.

Figure 3. Combination of PRT1419 and PRT3789 Demonstrates Potent Anti-tumor Activity Including Tumor Regressions in SMARCA4-Deleted Lung Cancer Models *In Vivo*

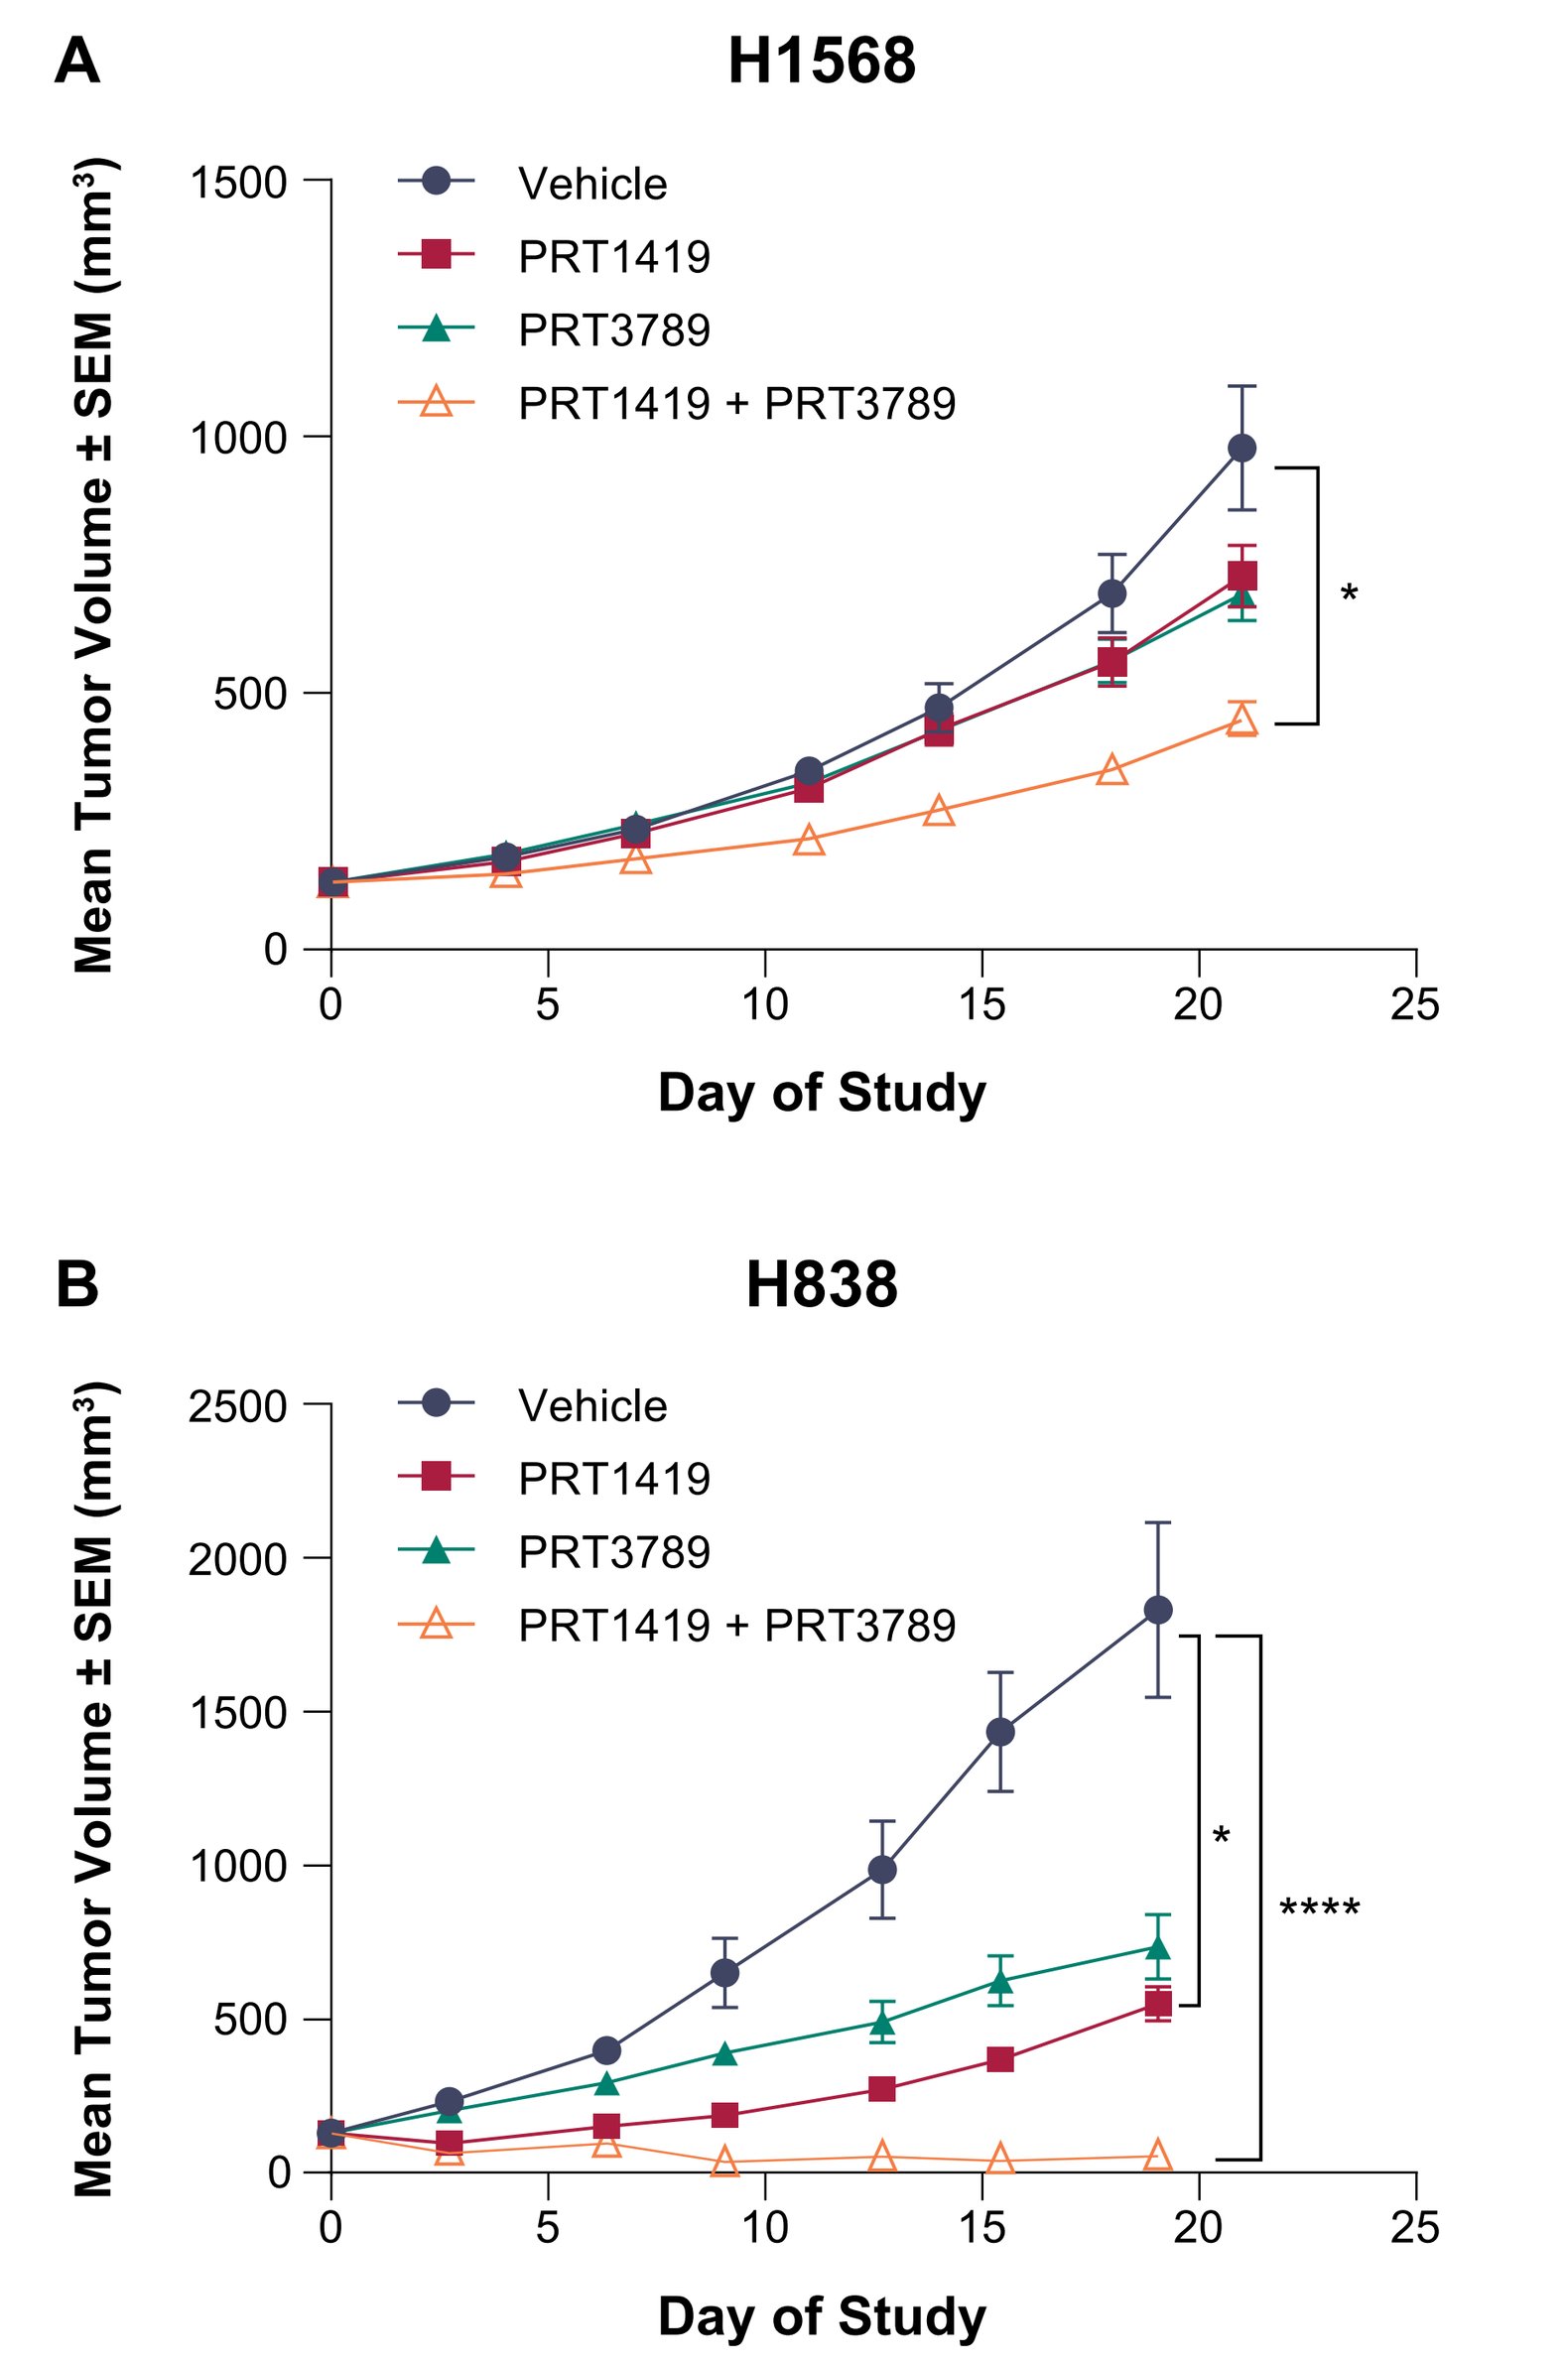
A
H1568
Vehicle
PRT1419
PRT3789
PRT1419 + PRT3789
Mean Tumor Volume ± SEM (mm³)
Day of Study
*
B
H838
Vehicle
PRT1419
PRT3789
PRT1419 + PRT3789
Mean Tumor Volume ± SEM (mm³)
Day of Study
*

PRT1419 combines with PRT3789 to potently repress tumor growth in cell line–derived xenograft (CDX) models of SMARCA4-deleted non-small cell lung cancer. Female BALB/c nude mice were injected with **(A)** NCI-H1568 or **(B)** NCI-H838 subcutaneously in the right front flank region. Animals were dosed with PRT1419 once a week and/or PRT3789 every 3 days. Eight mice per group, *P<0.05, ****P<0.0001 versus vehicle by Mann-Whitney U test. SEM, standard error of the mean.

Figure 4. SMARCA4-Deleted Lung Cancer Patient–Derived Xenograft Cell Lines With Low SMARCA2 Levels Are Sensitive to MCL1 Inhibition

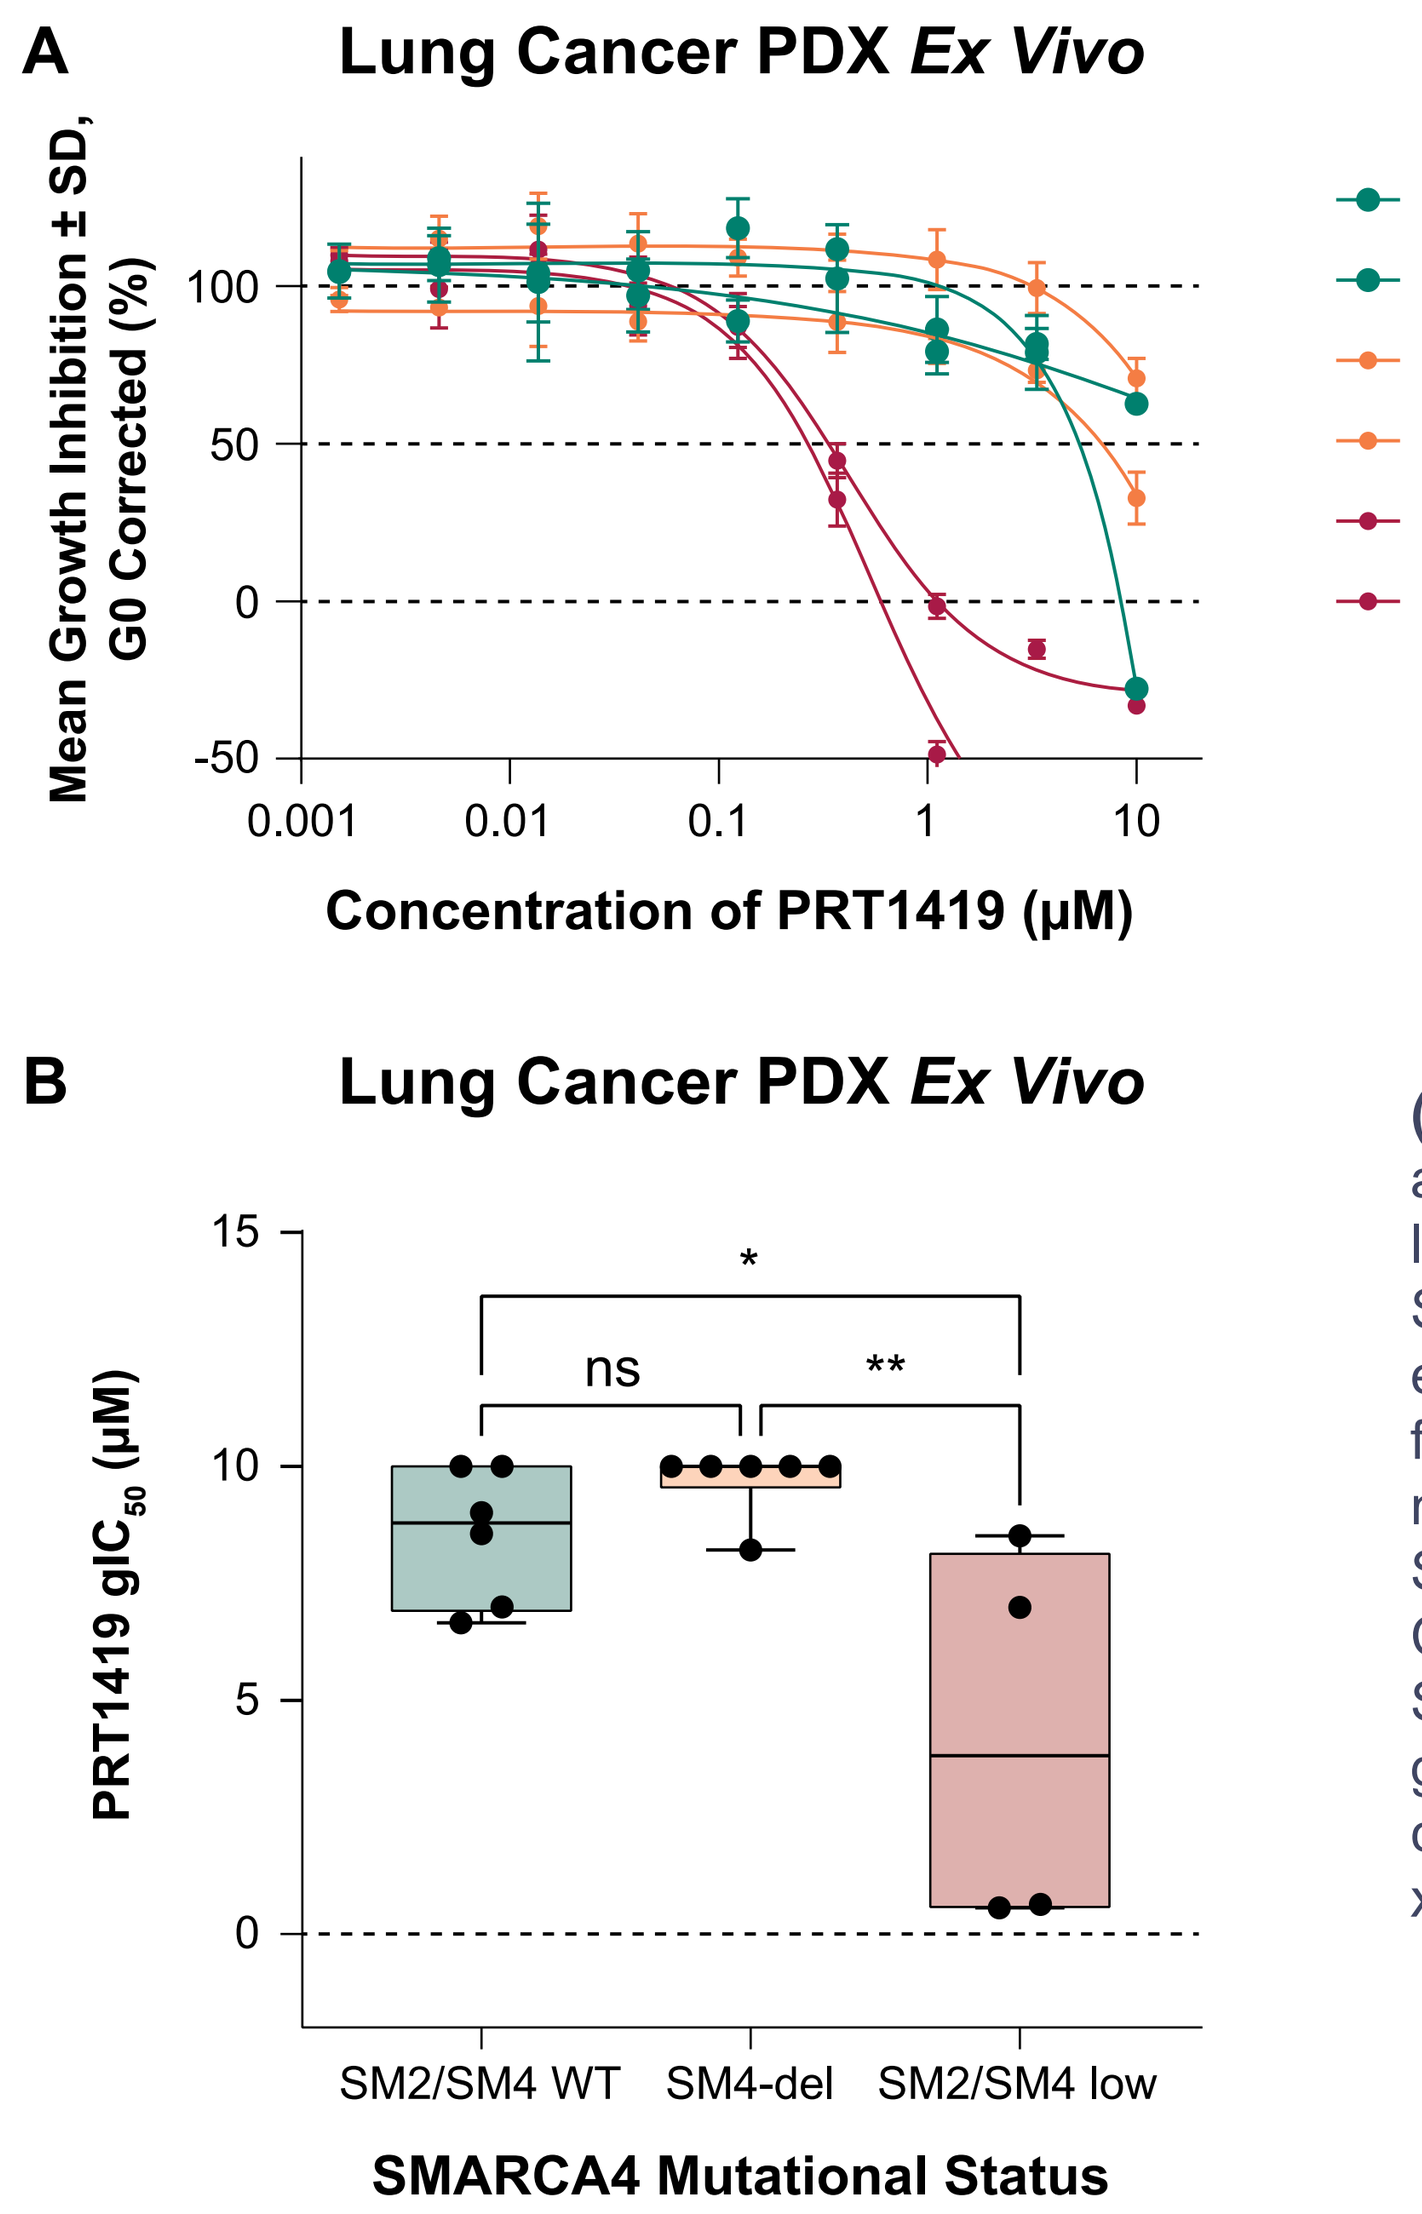
A
Lung Cancer PDX Ex Vivo
Mean Growth Inhibition ± SD, G0 Corrected (%)
Concentration of PRT1419 (μM)
LU2512 (SMARCA4 WT)
LU1147 (SMARCA4 WT)
LU2511 (SMARCA4 del)
LU1554 (SMARCA4 del)
LU5262 (SMARCA2/4 low)
LU6438 (SMARCA2/4 low)
B
Lung Cancer PDX Ex Vivo
PRT1419 gIC50 (μM)
*
ns
**
SM2/SM4 WT
SM4-del
SM2/SM4 low
SMARCA4 Mutational Status

(A, B) 3D cell viability assay assessing PRT1419 sensitivity in lung PDX models of varying SMARCA2/4 status. PDX lines were established from tumors harvested from live animals and cultured in a methylcellulose-based 3D assay. Sensitivity to PRT1419 was assessed by CellTiter-glo. *P<0.05, **P<0.01 versus SM2/SM4 WT by one-way ANOVA. gIC_{50}, growth half-maximal inhibitory concentration; PDX, patient-derived xenograft.

Figure 5. SMARCA2 Degradation Modulates Expression of Key Apoptosis Mediators

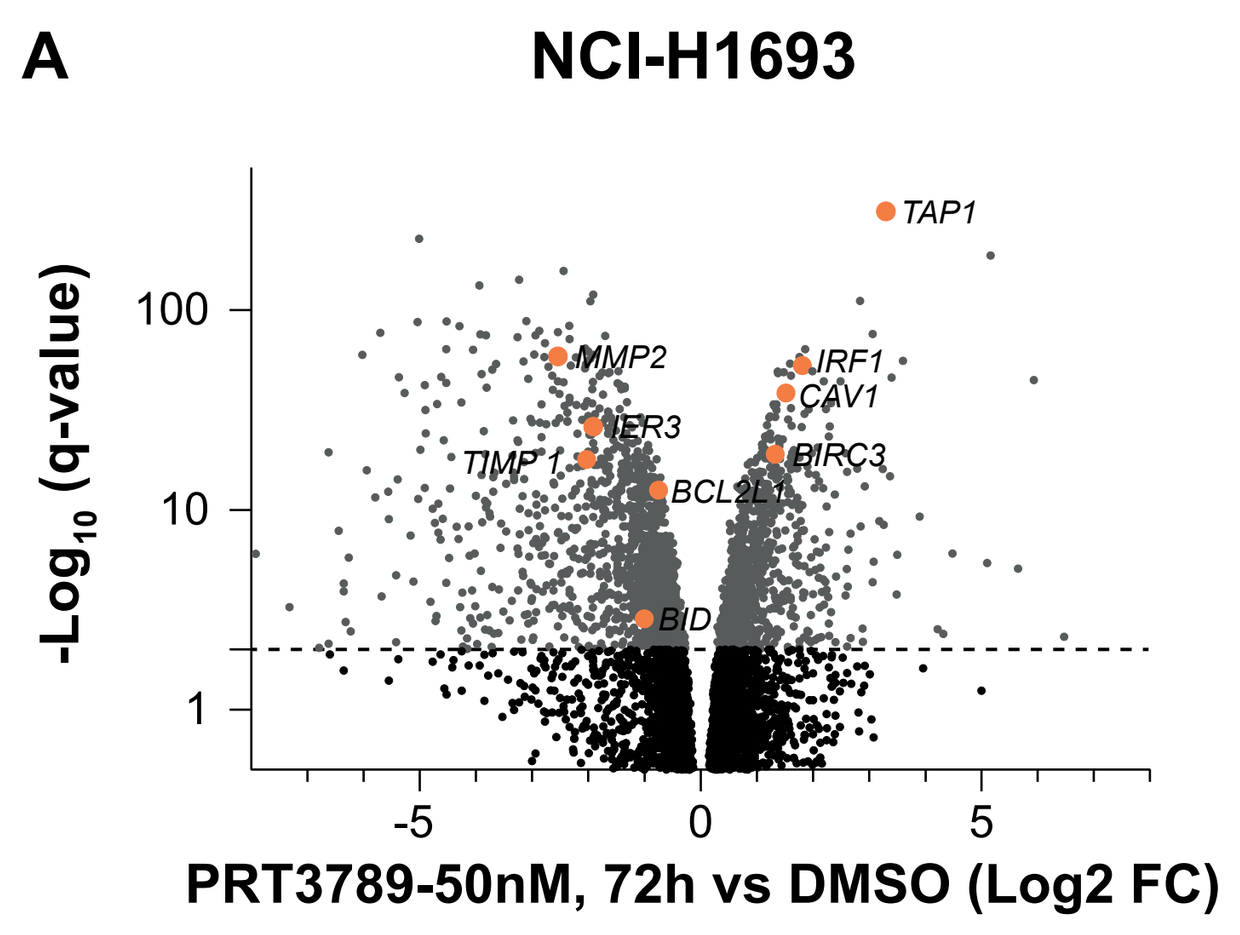
A
NCI-H1693
-Log10 (q-value)
TAP1
MMP2
IRF1
CAV1
IER3
TIMP1
BIRC3
BCL2L1
BID
PRT3789-50nM, 72h vs DMSO (Log2 FC)

B

Gene	Description
TAP1	ATP-binding cassette transporter; promotes apoptosis in mesothelioma[4]
BCL2L1	Anti-apoptotic member of the BCL2 family
BID	Pro-apoptotic member of the BCL2 family
TIMP1	Metalloproteinase inhibitor; binds BCL2 to inhibit apoptosis[5]
IER3	Stress-inducible gene; interacts with BCL2 proteins to inhibit apoptosis[6]
MMP2	MMP; promotes apoptosis via N-cadherin cleavage[7]
IRF1	Transcription factor; induces apoptosis via transcription of BAK and caspases[8]
CAV1	Structural protein; inhibits apoptosis via activation of PI3K/AKT[9]
BIRC3	Member of the inhibitors of apoptosis family[10]

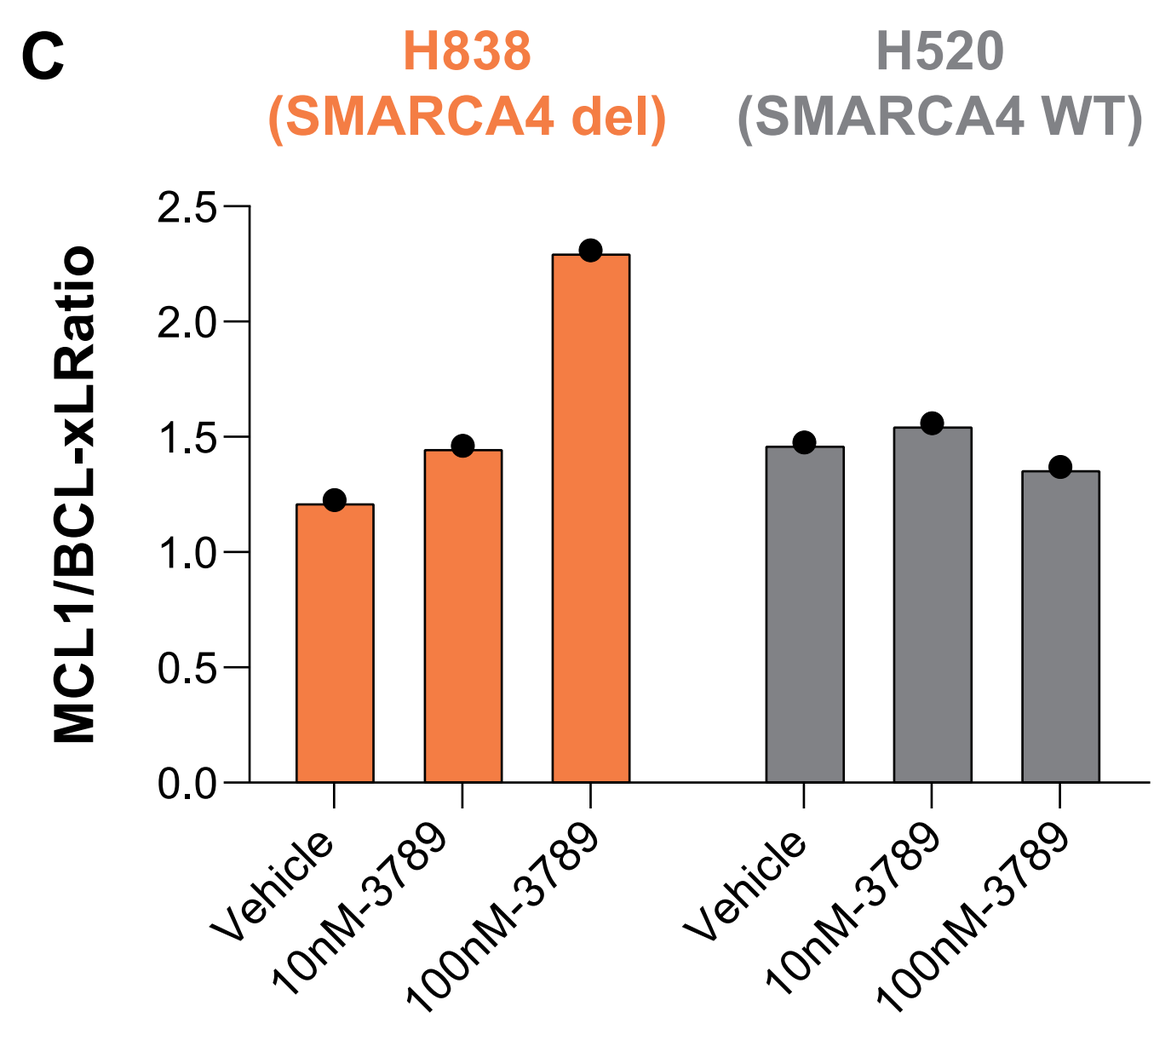
C
H838 (SMARCA4 del)
H520 (SMARCA4 WT)
MCL1/BCL-xLRatio
Vehicle
10nM-3789
100nM-3789
Vehicle
10nM-3789
100nM-3789

(A) Volcano plots of global RNA expression data in PRT3789-treated NCI-H1693 (SMARCA4-del); genes mediating apoptosis are highlighted in orange. Unbiased pathway enrichment analysis on differentially expressed genes using the Data4Cure platform (https://data4cure.com) identified apoptosis as an enriched pathway following PRT3789 treatment (BI score=0.44; P=0.0002; Q=0.001). **(B)** Key apoptosis mediators deregulated by PRT3789. **(C)** Western blot assessing MCL1/BCLxL ratio in a SMARCA4-del and SMARCA4 WT cell line following treatment with PRT3789. MCL1/BCLxL ratio computed as ratio of band intensities following quantification with ImageJ, n = 1. AKT, protein kinase B; BIRC3, baculoviral inhibitors of apoptosis repeat containing 3; CAV1, Caveolin 1; IRF1, Interferon Regulatory Factor 1; MMP, matrix metalloproteinase; PI3K, phosphoinositide 3-kinase; TAP1, transporter associated with antigen processing 1; TIMP1, tissue inhibitor matrix metalloproteinase 1.

Figure 6. Additional Co-occurring Alterations in SWI/SNF Factors Are Associated With PRT1419 Sensitivity *In Vitro*

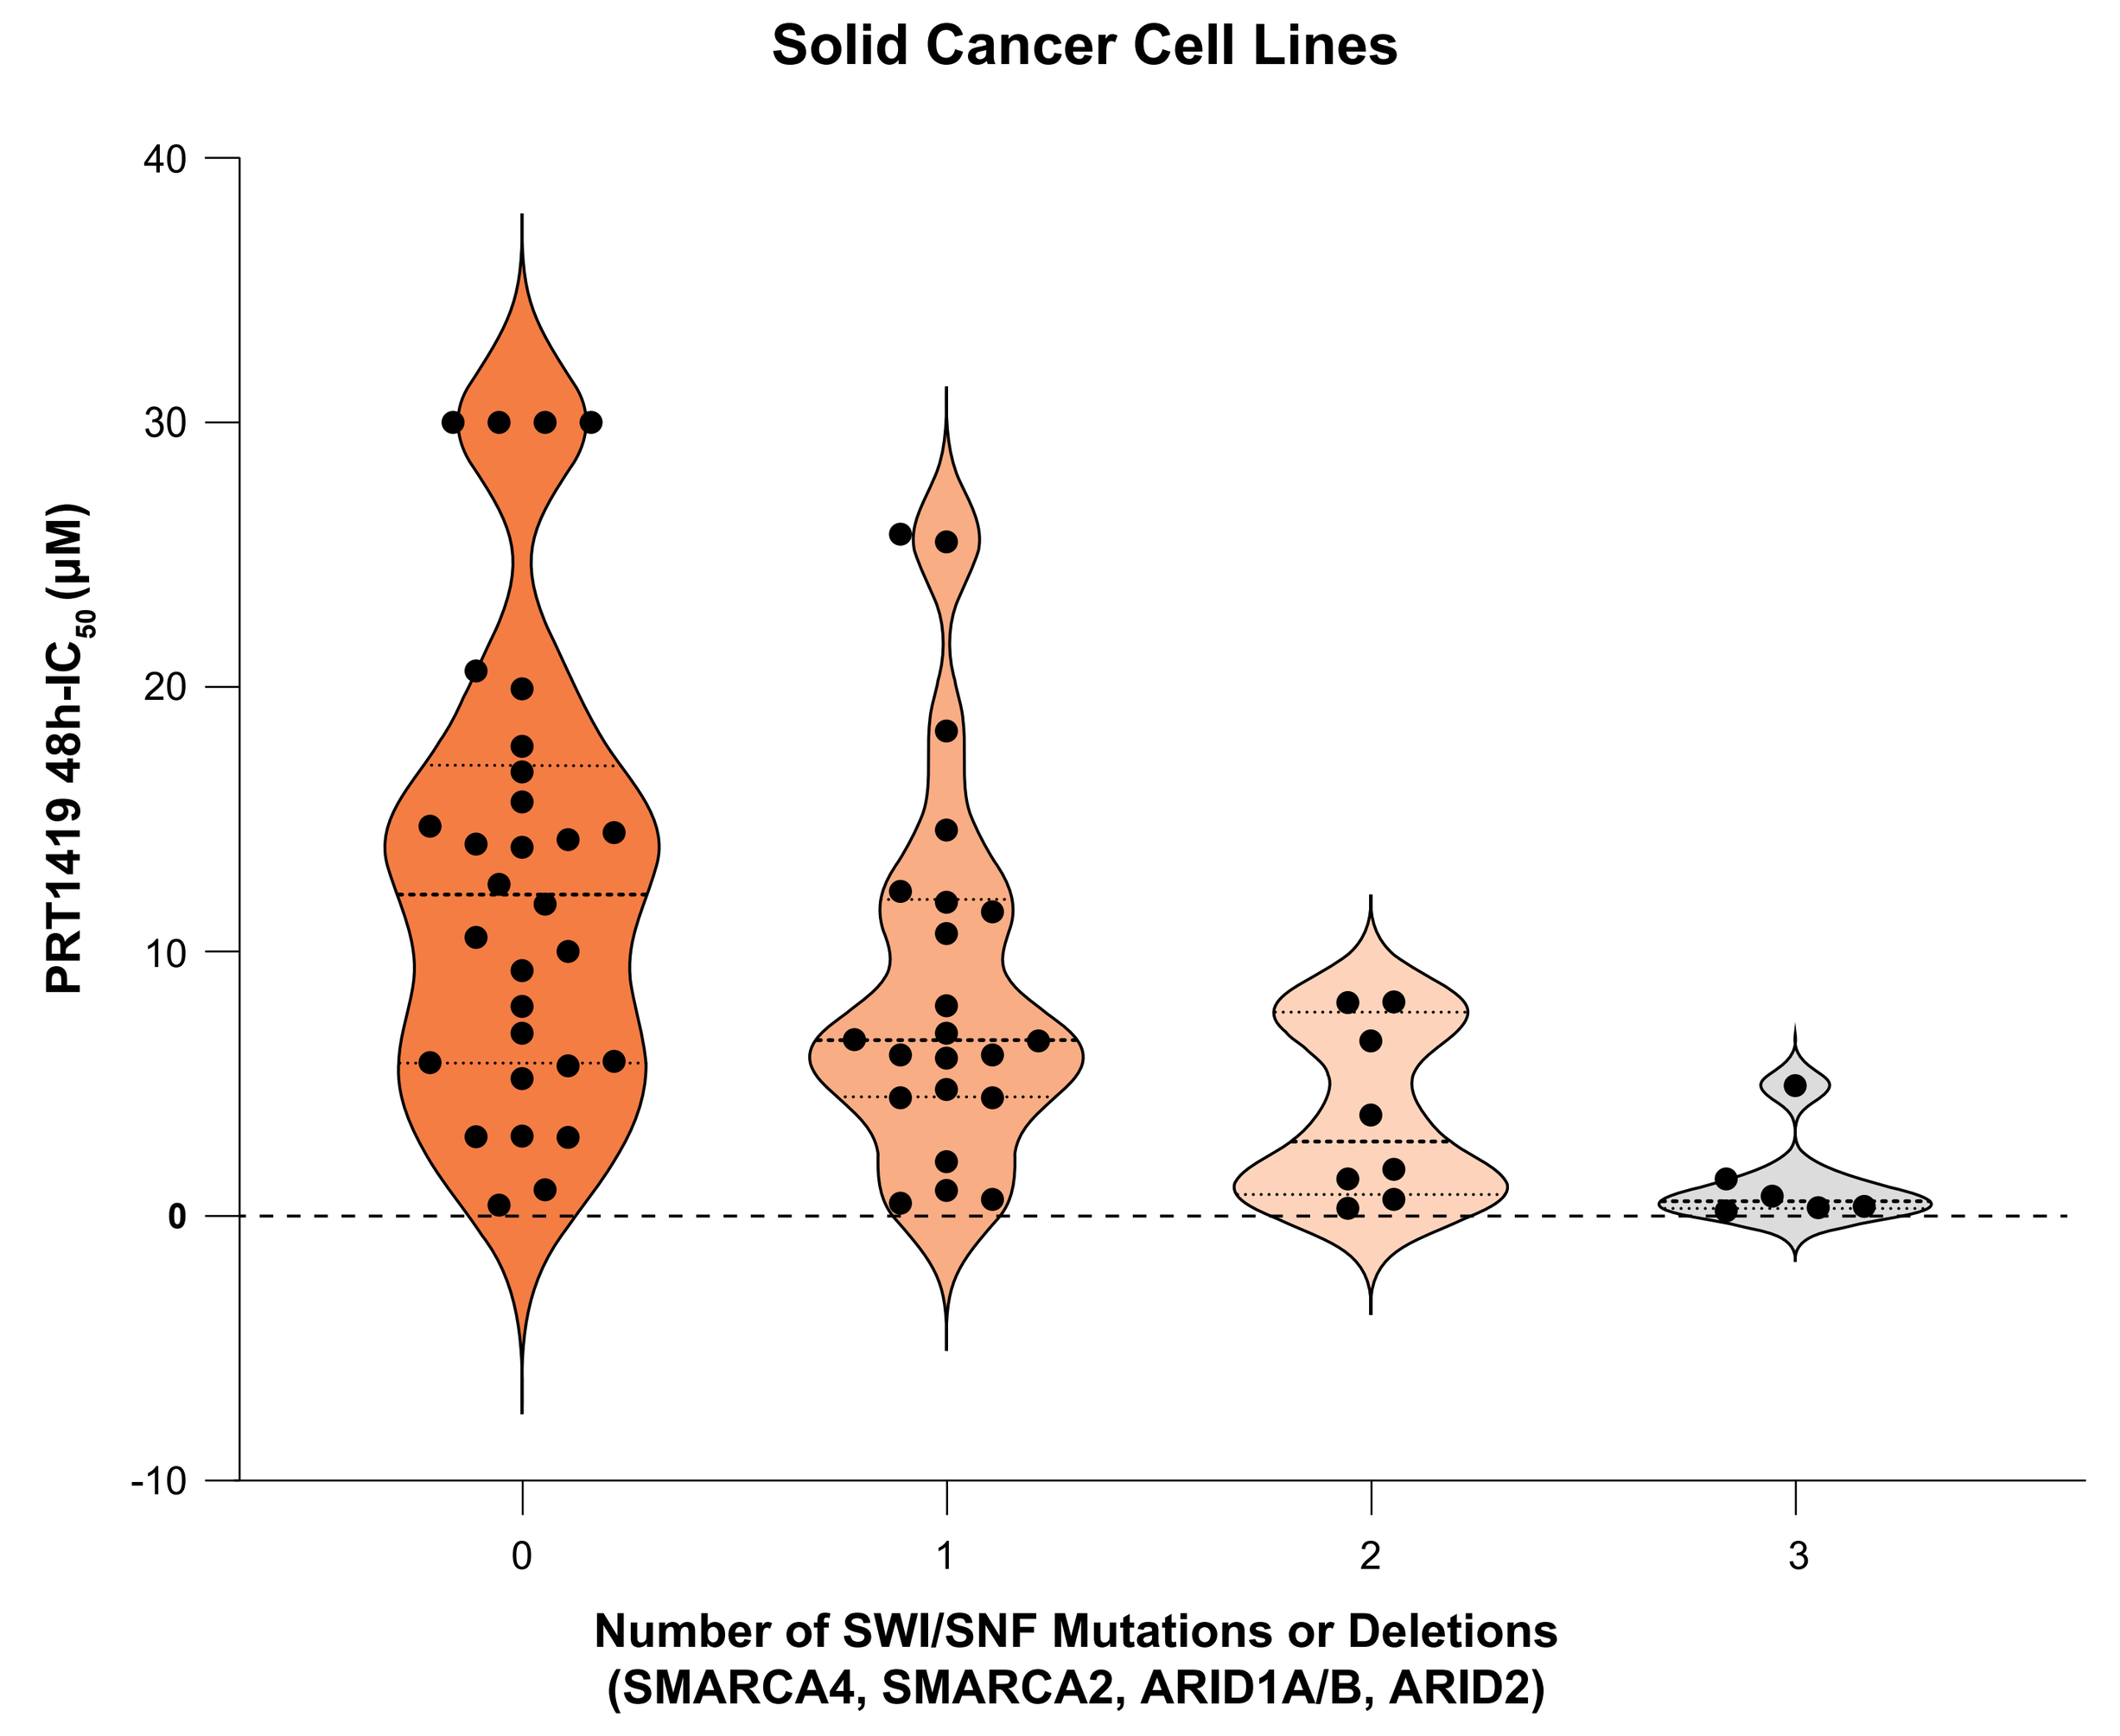
Solid Cancer Cell Lines
PRT1419 48h-IC50 (μM)
Number of SWI/SNF Mutations or Deletions
(SMARCA4, SMARCA2, ARID1A/B, ARID2)

PRT1419 IC_{50} assessed by CellTiter-Glo in a variety of human solid cancer lines, correlates with number of SWI/SNF alterations. Mutation data were retrieved from DepMap (https://depmap.org/portal/). IC_{50}, half-maximal inhibitory concentration.

Conclusions

- SMARCA2 degradation synergizes with MCL1 inhibition in preclinical models of SMARCA4-deleted lung cancer.
- SMARCA2 degradation modulates the expression of key apoptosis regulators sensitizing cells to MCL1 inhibition.
- Additional co-occurring mutations and alterations in SWI/SNF complex factors are associated with increased sensitivity to MCL1 inhibition.
- Preclinical studies evaluating PRT1419 in other tumor types with SWI/SNF mutations are ongoing.

References

1. Campbell KJ, et al. *Open Biol*. 2018;8:180002.
2. Hulse M, et al. Presented at AACR 2022, April 8-13, 2022, New Orleans, LA, USA. Poster 3263.
3. Rago F, et al. *Biochem Biophys Res Commun*. 2019;508:109-116.
4. Neuzil J, et al. *Biochem Biophys Res Commun*. 2006;343:1113-1117.
5. Nalluri S, et al. *PLoS One*. 2015;10:e0137673.
6. Ortega MM, et al. *J Clin Oncol*. 2008;26(15_suppl):8598.
7. Hartland SN, et al. *Liver Int*. 2009;29:966-978.
8. Kim PKM, et al. *Oncogene*. 2004;23:1125-1135.
9. Han F, et al. *Int J Clin Exp Pathol*. 2015;8:8937-8947.
10. Gressot LV, et al. *Oncotarget*. 2017;8:12695-12704.

Disclosures

All authors are employees of Prelude Therapeutics, Inc., Wilmington, DE and may own stocks or shares.

Editorial support was provided by Russell Craddock, PhD, of Parexel International, Uxbridge, UK, and was funded by Prelude Therapeutics Inc.

Copies obtained through the Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author.