upliFT-D: A Clinical Trial of Gene Therapy (PBFT02) for Frontotemporal Dementia With Progranulin Gene Mutations (FTD-*GRN*)

Voss T,[1] Grossman M,[2] Irwin DJ,[2] Schulz PE,[3] de Souza LC,[4] Triglia P,[1] Quadrini KJ,[1] Nagilla P,[1] Forman MS[1]

[1]Passage Bio, Inc., Philadelphia, PA, United States; [2]Department of Neurology, Perelman School of Medicine at the University of Pennsylvania; University of Pennsylvania Health System, Philadelphia, PA, United States; [3]Department of Neurology at McGovern Medical School, UTHealth Houston, Houston, TX, United States; [4]Neuropsychiatric Branch, Neurology Division, University Hospital, Universidade Federal de Minas Gerais, Belo Horizonte, Brazil

Background

- In up to 10% of patients with **frontotemporal dementia (FTD)**, the disease is caused by changes or mutations in a gene called ***GRN*** that makes a protein called **progranulin (PGRN)**.[1]
- Most *GRN* mutations lead to insufficient PGRN in the brain to ensure proper function.[2]
- To increase the levels of PGRN in the brain, a potential new gene therapy called **PBFT02** has been developed by Passage Bio, Inc.[3,4]
- PBFT02 is a one-time gene therapy that delivers a healthy working copy of the *GRN* gene to the brain, which then makes more PGRN, reducing or eliminating the protein deficiency in patients with FTD-*GRN*.[3,5]
- Gene therapy offers a way to deliver a healthy working copy of the gene using an envelope called the **adeno-associated virus (AAV) vector** (a way to deliver genetic material into cells).[4]

Preclinical Evidence

- Passage Bio tested several AAV vectors for *GRN* gene delivery and selected the one that led to the greatest increase in PGRN levels (**Figure 1**).[6]
- When PBFT02 was tested in monkeys it was shown that the delivery of a functional human *GRN* gene to the brain resulted in a substantial increase of PGRN levels in cerebrospinal fluid surrounding the brain[6] (**Figure 1**, **red arrows**).

Figure 1 Production of PGRN Following the Administration of AAV Vectors Carrying the Human *GRN* Gene to the Monkey Brain[6]

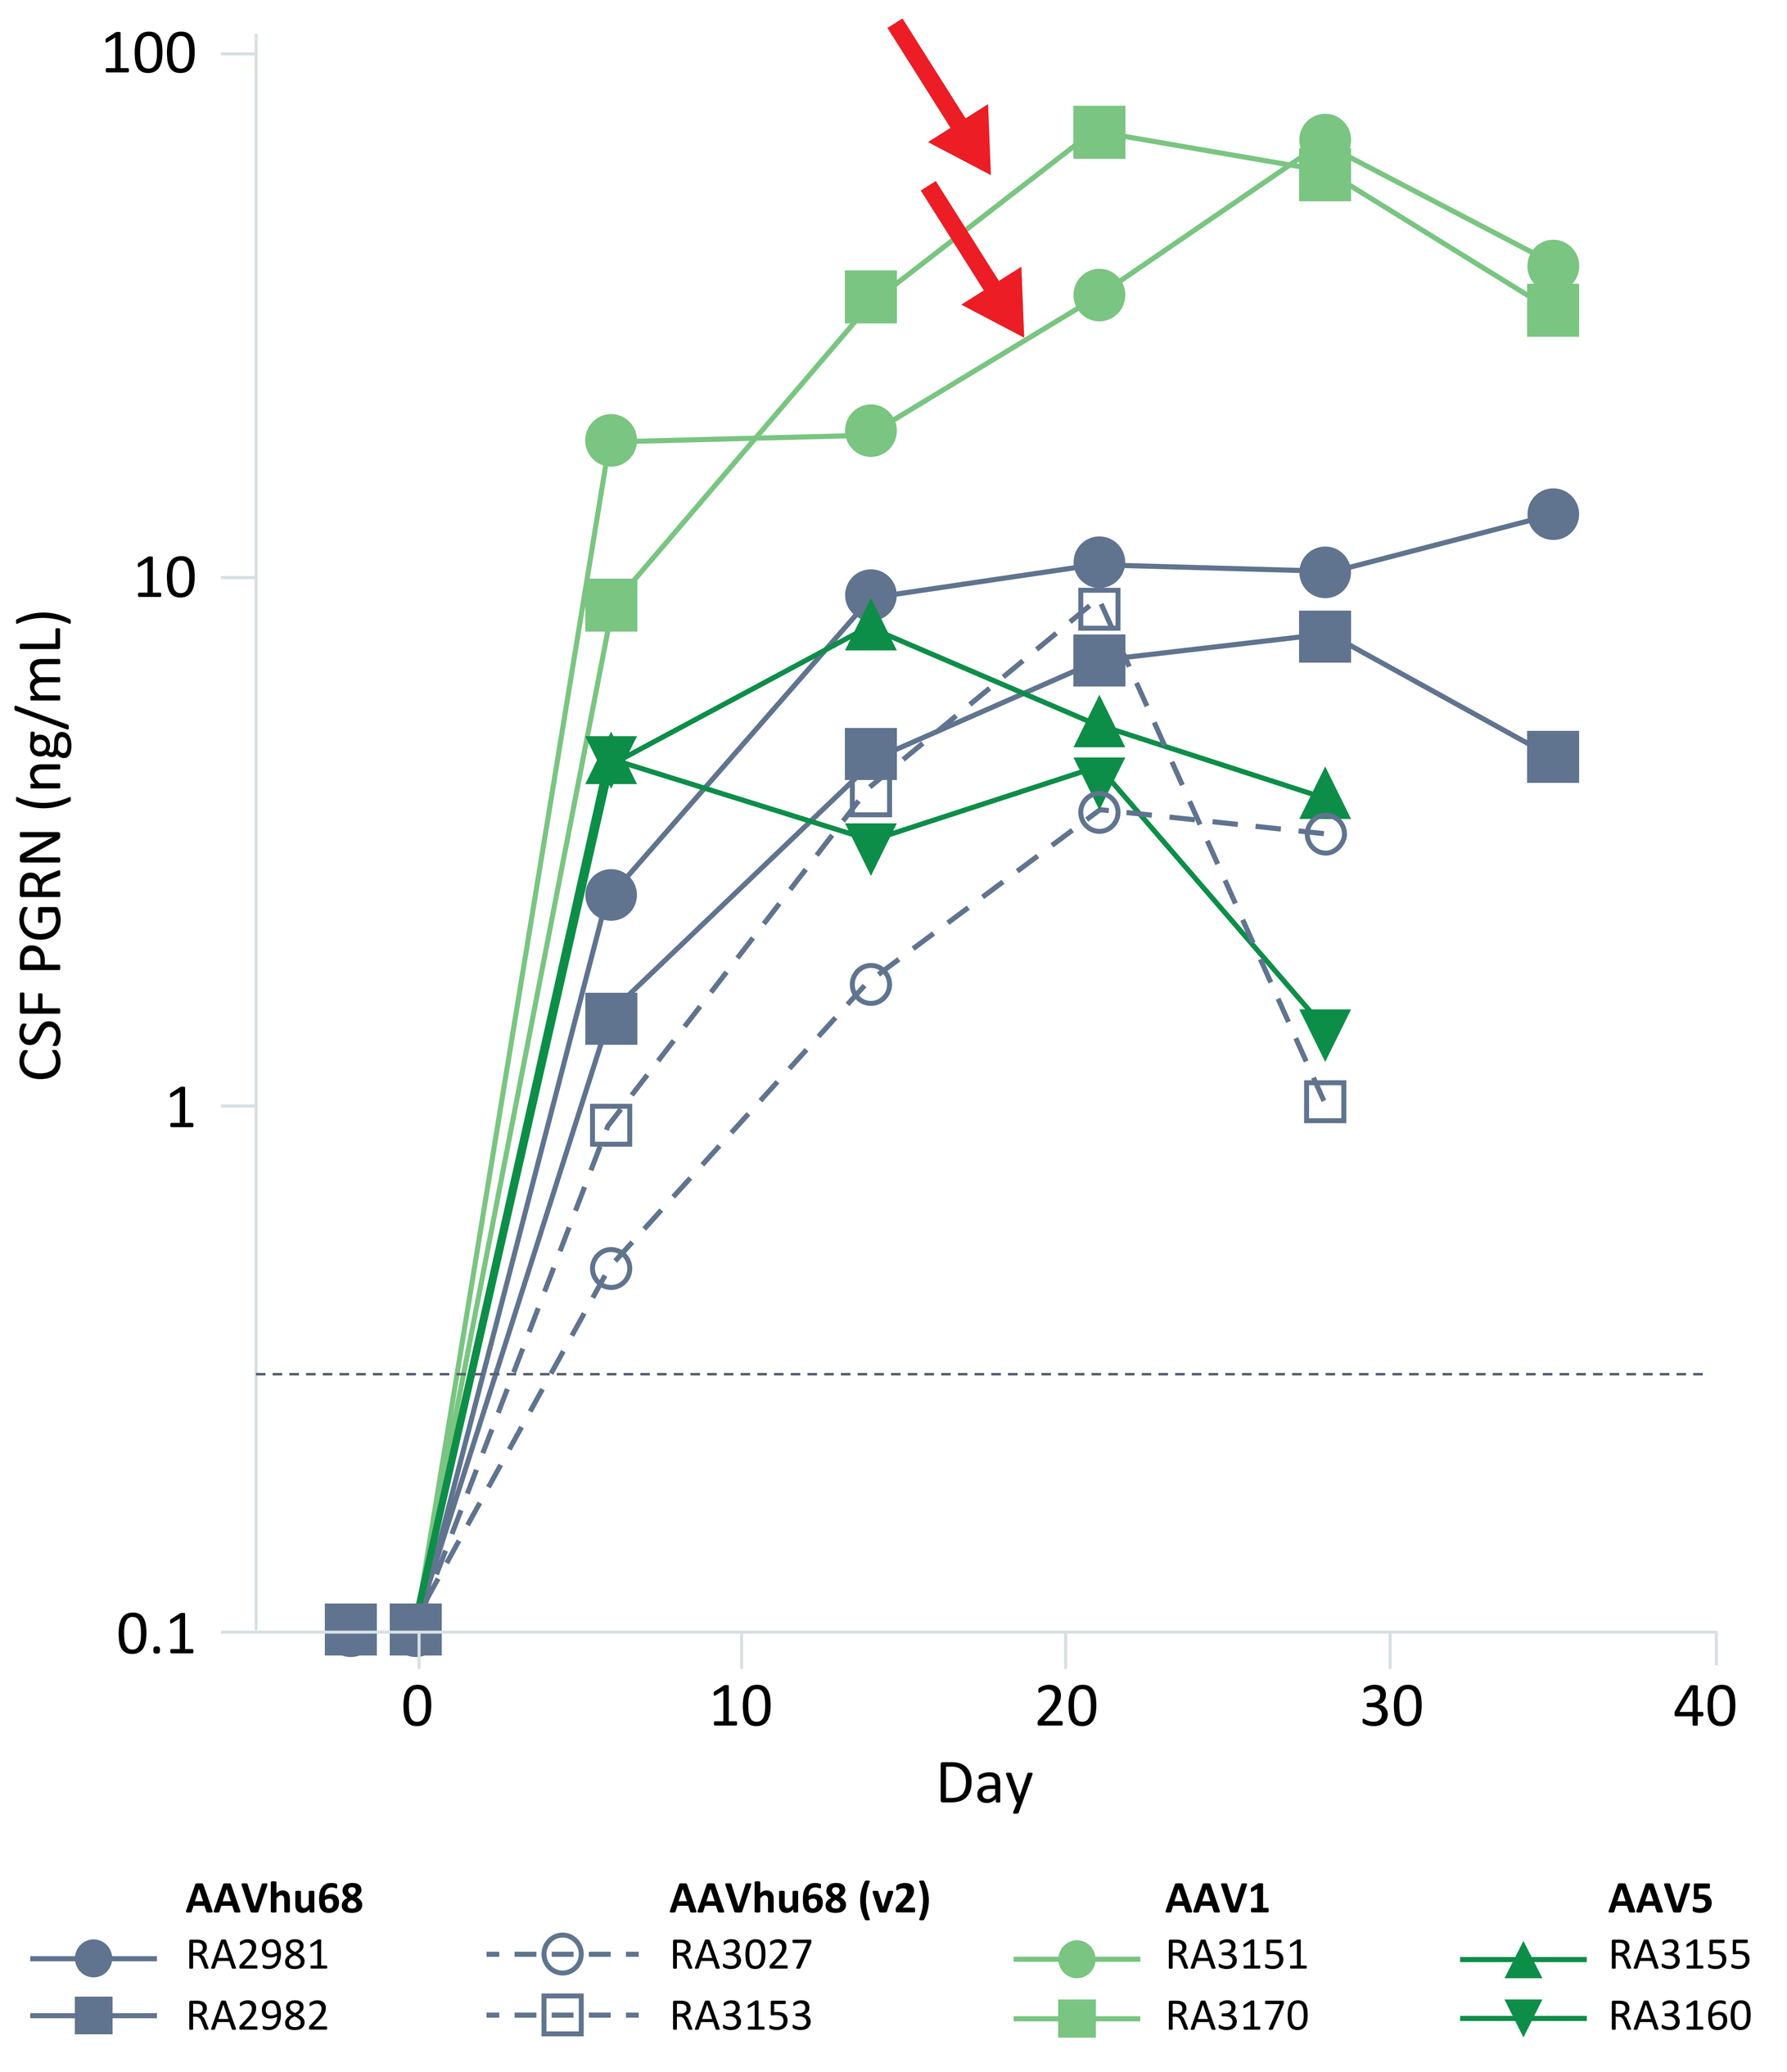

CSF PGRN (ng/mL)
100
10
1
0.1
0
10
20
30
40
Day
AAVhu68
RA2981
RA2982
AAVhu68 (v2)
RA3027
RA3153
AAV1
RA3151
RA3170
AAV5
RA3155
RA3160

Additional Information for Patients and Caregivers

- Patients with FTD may be eligible for enrollment in clinical trials of investigative therapies, so an early diagnosis is critical. Caregivers should reach out to their health care professional to learn more.

Links to Resources

- If you would like to learn more about FTD or the upliFT-D clinical trial, please visit: ftdclinicaltrial.com.
- Please contact Passage Bio to learn more about the upliFT-D clinical trial: https://passagebioclinicaltrials.com/Frontotemporal-Dementia/#contactForm.

upliFT-D – A First-in-Human Clinical Trial

Trial Design

- **All patients** will receive **treatment** in the upliFT-D trial (**Figure 2**).[5,7]
- **Cohort 1**: patients will receive a low dose of PBFT02; **Cohort 2**: patients will receive a higher dose of PBFT02; and **Cohort 3**: an optional third dose level may be studied, depending on the results from the first 2 groups.[5,7]

Figure 2 The upliFT-D Trial Design[5,7]

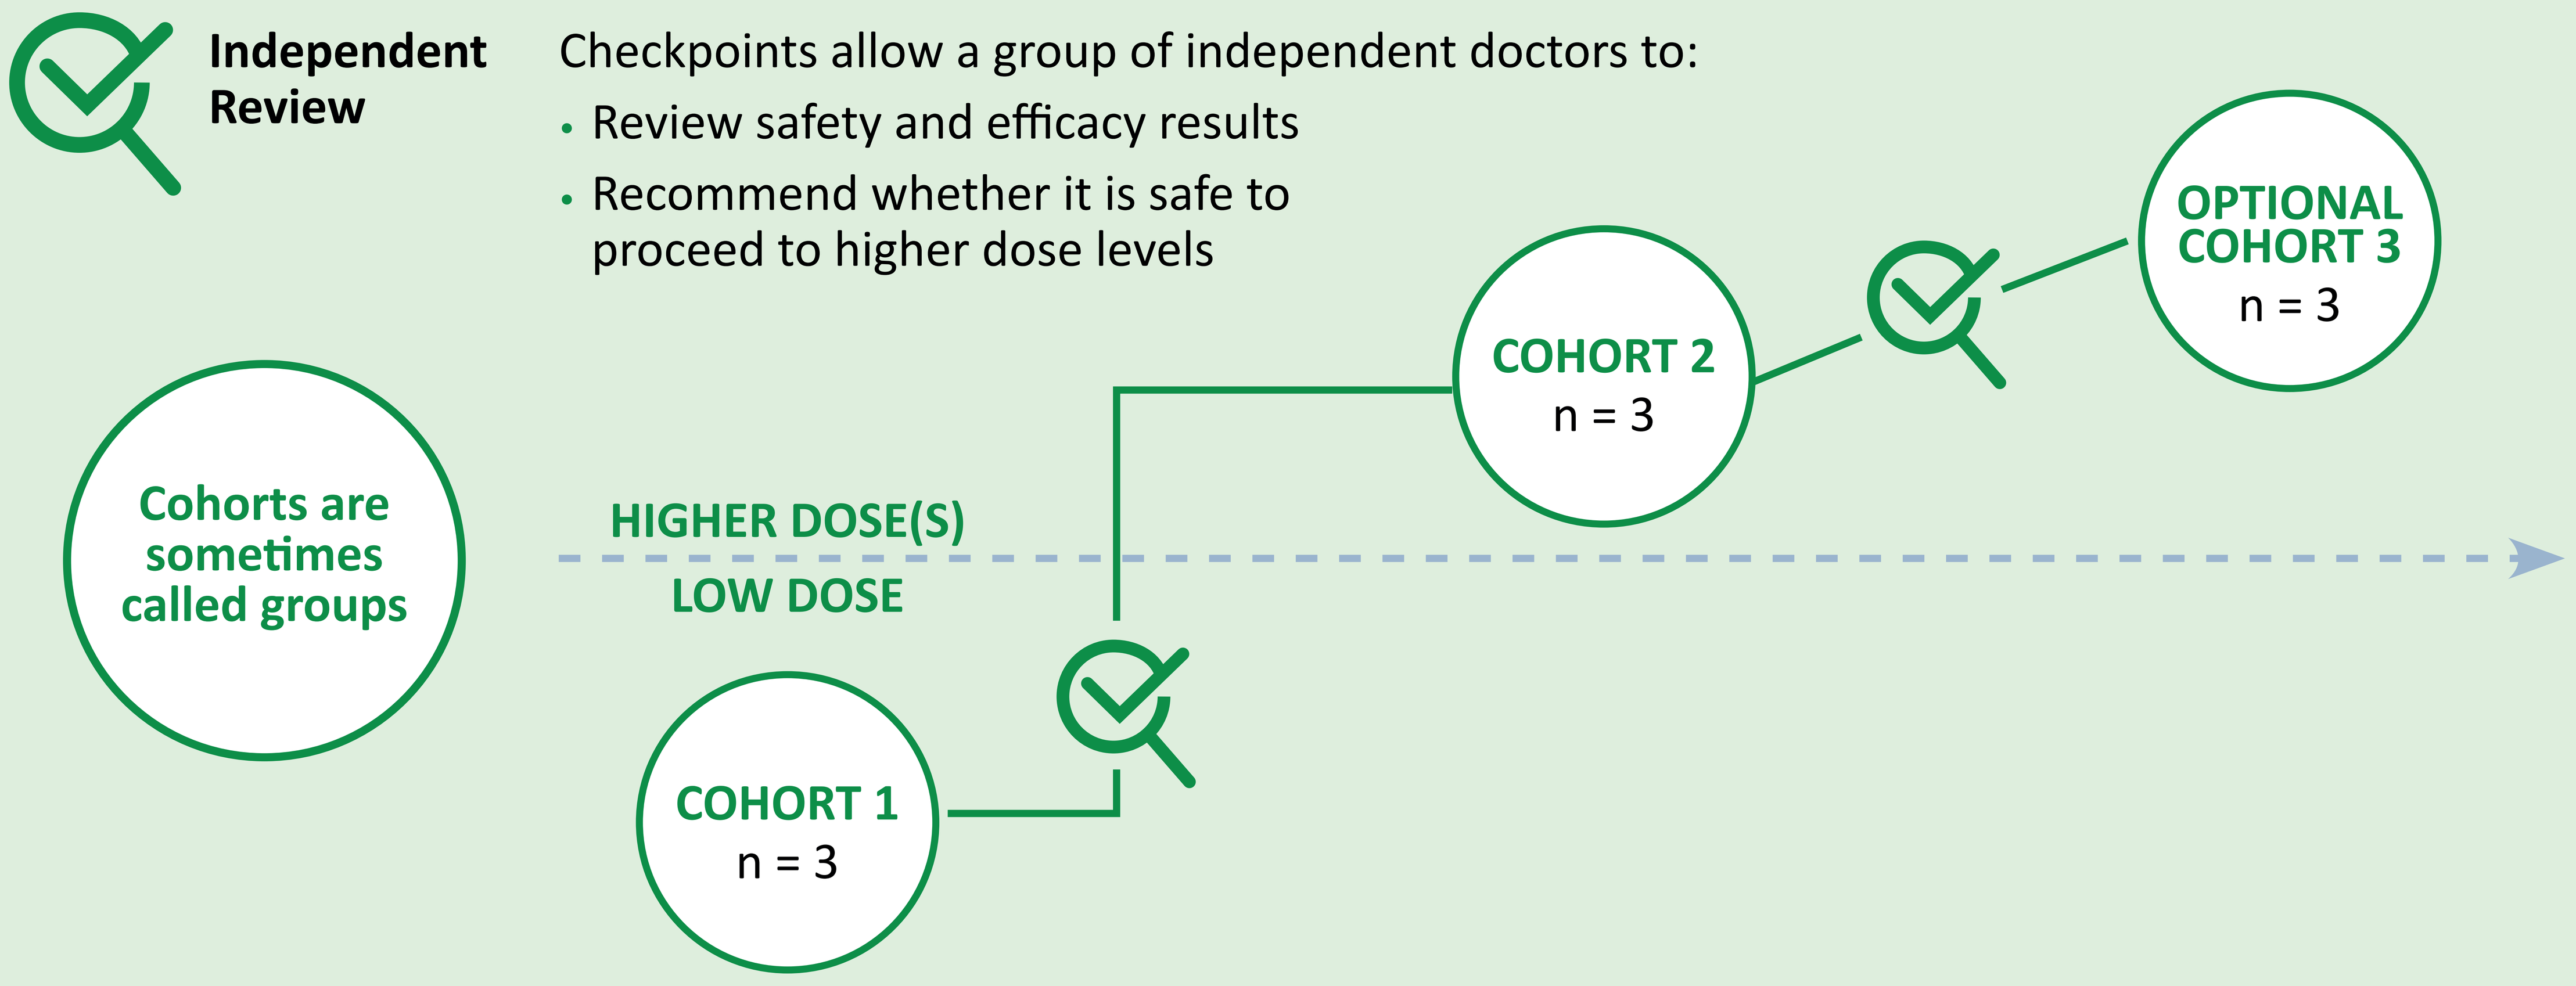

Independent Review
Checkpoints allow a group of independent doctors to:
• Review safety and efficacy results
• Recommend whether it is safe to proceed to higher dose levels
Cohorts are sometimes called groups
HIGHER DOSE(S)
LOW DOSE
COHORT 1 n = 3
COHORT 2 n = 3
OPTIONAL COHORT 3 n = 3

- Patients will be followed for up to 5 years, including a 2-year main study and a 3-year safety extension study (**Figure 3**).[8]

Figure 3 Treatment Duration[8]

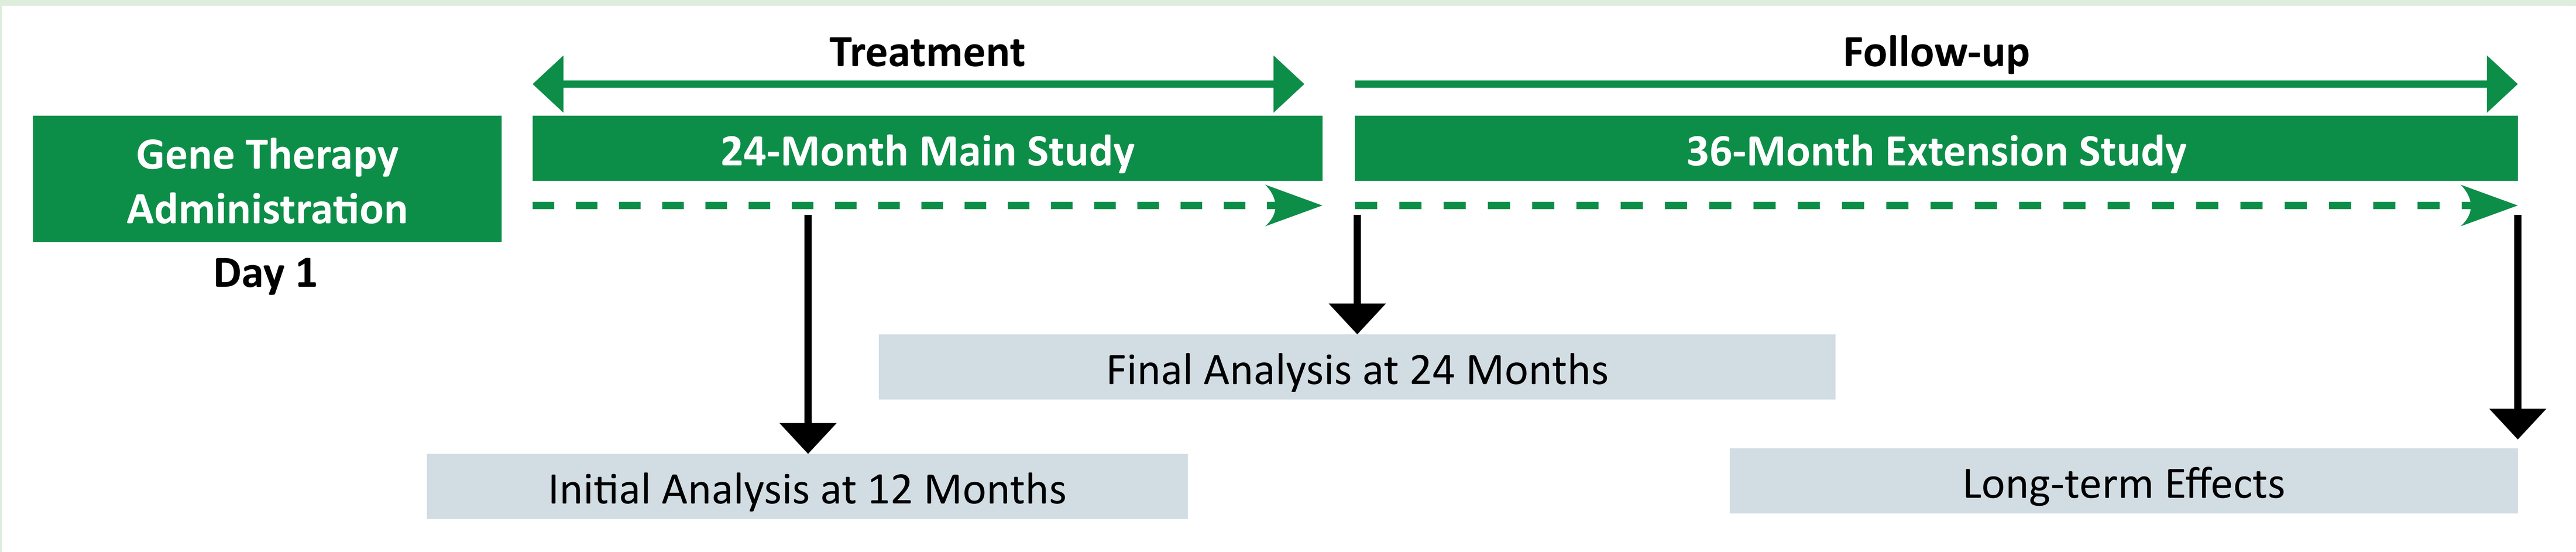

Treatment
Follow-up
Gene Therapy Administration
Day 1
24-Month Main Study
36-Month Extension Study
Final Analysis at 24 Months
Initial Analysis at 12 Months
Long-term Effects

Goals of the Clinical Trial

- The upliFT-D clinical trial[5] is designed to provide information about the safety, effectiveness, and appropriate dosing of PBFT02 gene therapy in patients with FTD-*GRN* (**Figure 4**).[7]
- Safety and efficacy will be monitored with different types of tests.

Figure 4 Data Collection in the upliFT-D Clinical Trial[5,7]

SAFETY

Is the treatment…
- Safe?
- Causing side effects? If so, what are they?

EFFECTIVENESS

Does the treatment…
- Help improve cognitive functions?
- Help improve behavioral and language symptoms?

DOSING

Do different doses…
- Change the effectiveness of the potential treatment?
- Change the type and intensity of side effects?

Key Eligibility Criteria[5,7]

- To be eligible to join this clinical trial, participants must:
 - be aged **35 to 75 years**
 - have a diagnosis of FTD with **a confirmed *GRN* mutation**
 - have **mild FTD symptoms**
 - have **no complicating medical conditions** and
 - be living in the community.
- There are other criteria that must be met in order to join this clinical trial.[5]

How will gene therapy be administered?

- Participants will receive a **single injection** of the PBFT02 gene therapy into a region called the **cisterna magna**,[5,7] which is the space between the brain and the spinal cord; the injection will be administered by an interventional radiologist or a neurosurgeon.[9]

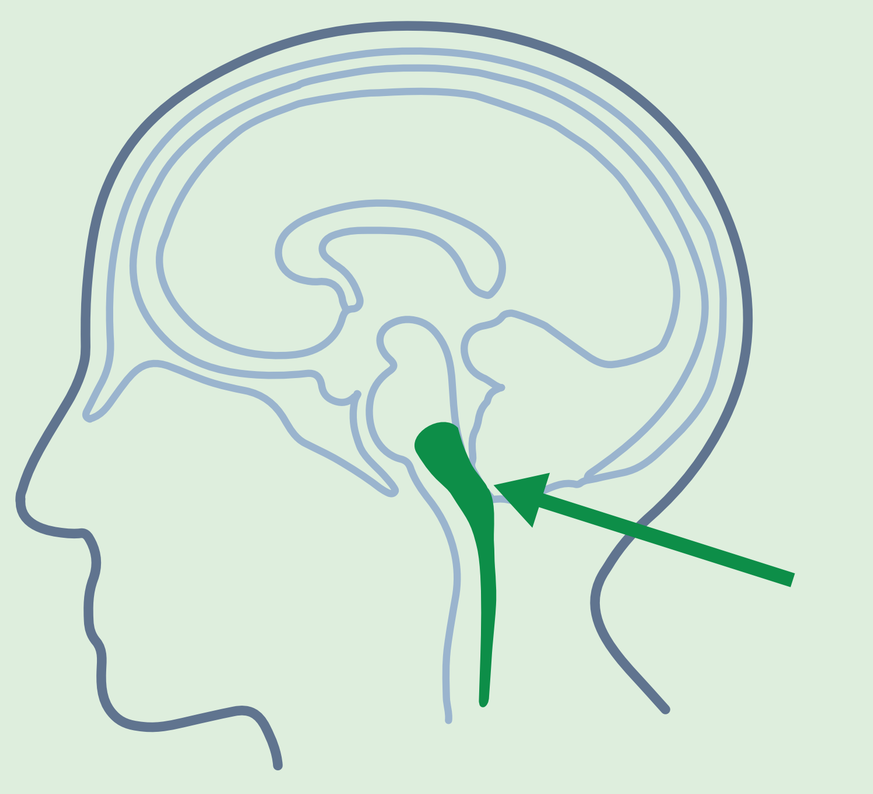

References

1. Gijselinck I, et al. *Hum Mutat*. 2008;29(12):1373–1386.
2. Rhinn H, et al. *Trends Pharmacol Sci*. 2022;43(8):641–652.
3. Passage Bio, Inc. Annual Report. 2020.
4. Naso MF, et al. *BioDrugs*. 2017;31(4):317–334.
5. Clinicaltrials.gov (NCT04747431); https://www.clinicaltrials.gov/ct2/show/NCT04747431?term=NCT04747431&draw=2&rank=1. Accessed September 28, 2022.
6. Hinderer C, et al. *Ann Clin Transl Neurol*. 2020;7(10):1843–1853.
7. Passage Bio. About upliFTD Passage Bio Trial information. Accessed September 28, 2022.
8. Passage Bio. PBFT02 trial protocol.
9. Pukenas B, et al. *Mol Genet Metab*. 2020;132;S89.

Disclosures

MG is supported by NIH (AG066597, AG052943), Department of Defense, and foundation sources (Samuel Newhouse Foundation, Peisach Family Foundation), and currently participates in trials sponsored by Prevail, Biogen, and Life Molecular Imaging. **DJI** receives research support to conduct therapeutic trials from Alector and Prevail and is supported by grants from NIH grants R01-NS109260, P01-AG066597, P30-AG10124, U19-AG062418, and the Penn Institute on Aging. **PES** participates in trials with Lilly, Roche, UCB Pharma, Biogen, Passage Bio, Inc., and others and has been an advisor and speaker for Lilly, Roche, and Biogen. He is funded by a dozen NIH grants and several Foundations. **LCdS** has no conflicts of interest to disclose. **TV**, **PT**, **KJQ**, **PN**, and **MSF** are employees of Passage Bio, Inc. and have received stock options.

Copies of this poster presentation obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission of Passage Bio, Inc.

https://bit.ly/3SvWKuC

Passage Bio

Presented at ISFTD 2022, November 2–5, 2022, Paris/Lille, France

Contact: tvoss@passagebio.com

GS3534_273843_Passage Bio__ISFTD Caregiver Poster S06.indd 1

10/26/22 12:54 PM