MCL1 inhibitor PRT1419 Demonstrates Antitumor Activity in PBRM1-altered Clear Cell Renal Cancer and Synergizes with Standard of Care Agents

Prelude THERAPEUTICS

Norman Fultang[1], Ashley M. Schwab[1], Brian Vidal Torres[1], Alexander Grego[1], Stephanie Rodgers[1], Diane Heiser[1], Kris Vaddi[1], Neha Bhagwat[1], Peggy Scherle[1]

[1]Prelude Therapeutics Incorporated, Wilmington, DE; contact: nfultang@preludetx.com

6147

Background

- Induced myeloid leukemia cell differentiation protein (MCL1) is a member of the B-cell lymphoma-2 (BCL2) family of apoptosis regulators and plays a critical role in promoting cancer cell survival[1].
- We previously described PRT1419, a novel, potent, and orally bioavailable MCL1 inhibitor that demonstrates anti-tumor efficacy in various preclinical models and is currently under evaluation in phase 1 clinical trials in hematologic and solid malignancies.[2]
- Gene dependency analyses to identify biomarkers of MCL1 inhibitor sensitivity revealed that clear cell Renal Cancer (ccRCC) cell lines with deleterious alterations in PBRM1/BAF180 (Polybromo 1) displayed a strong dependency on MCL1. We had previously described alterations in other mammalian SWI/SNF factors as biomarkers of MCL1 inhibitor sensitivity.[3]
- PBRM1 is a chromatin-targeting subunit of mammalian pBAF (SWI/SNF-B) complexes, which is frequently altered in various human cancers with a particularly high alteration rate in ccRCC (~40% of tumors harbor damaging PBRM1 alterations).[4]
- Current standard of care (SoC) agents in ccRCC include multi-targeted Receptor Tyrosine Kinase inhibitors, mammalian Target of Rapamycin (mTORc) inhibitors, HIF2α inhibitors and immune checkpoint blockade.[5]

Key Findings

- Here we show that PBRM1-loss is associated with sensitivity to MCL1 inhibition in ccRCC and provide rationale for the evaluation of PRT1419 in combination with SoC agents for the treatment of PBRM1-deficient ccRCC

Results

Figure 1. Damaging Mutations in *PBRM1* are associated with MCL1 dependency in ccRCC

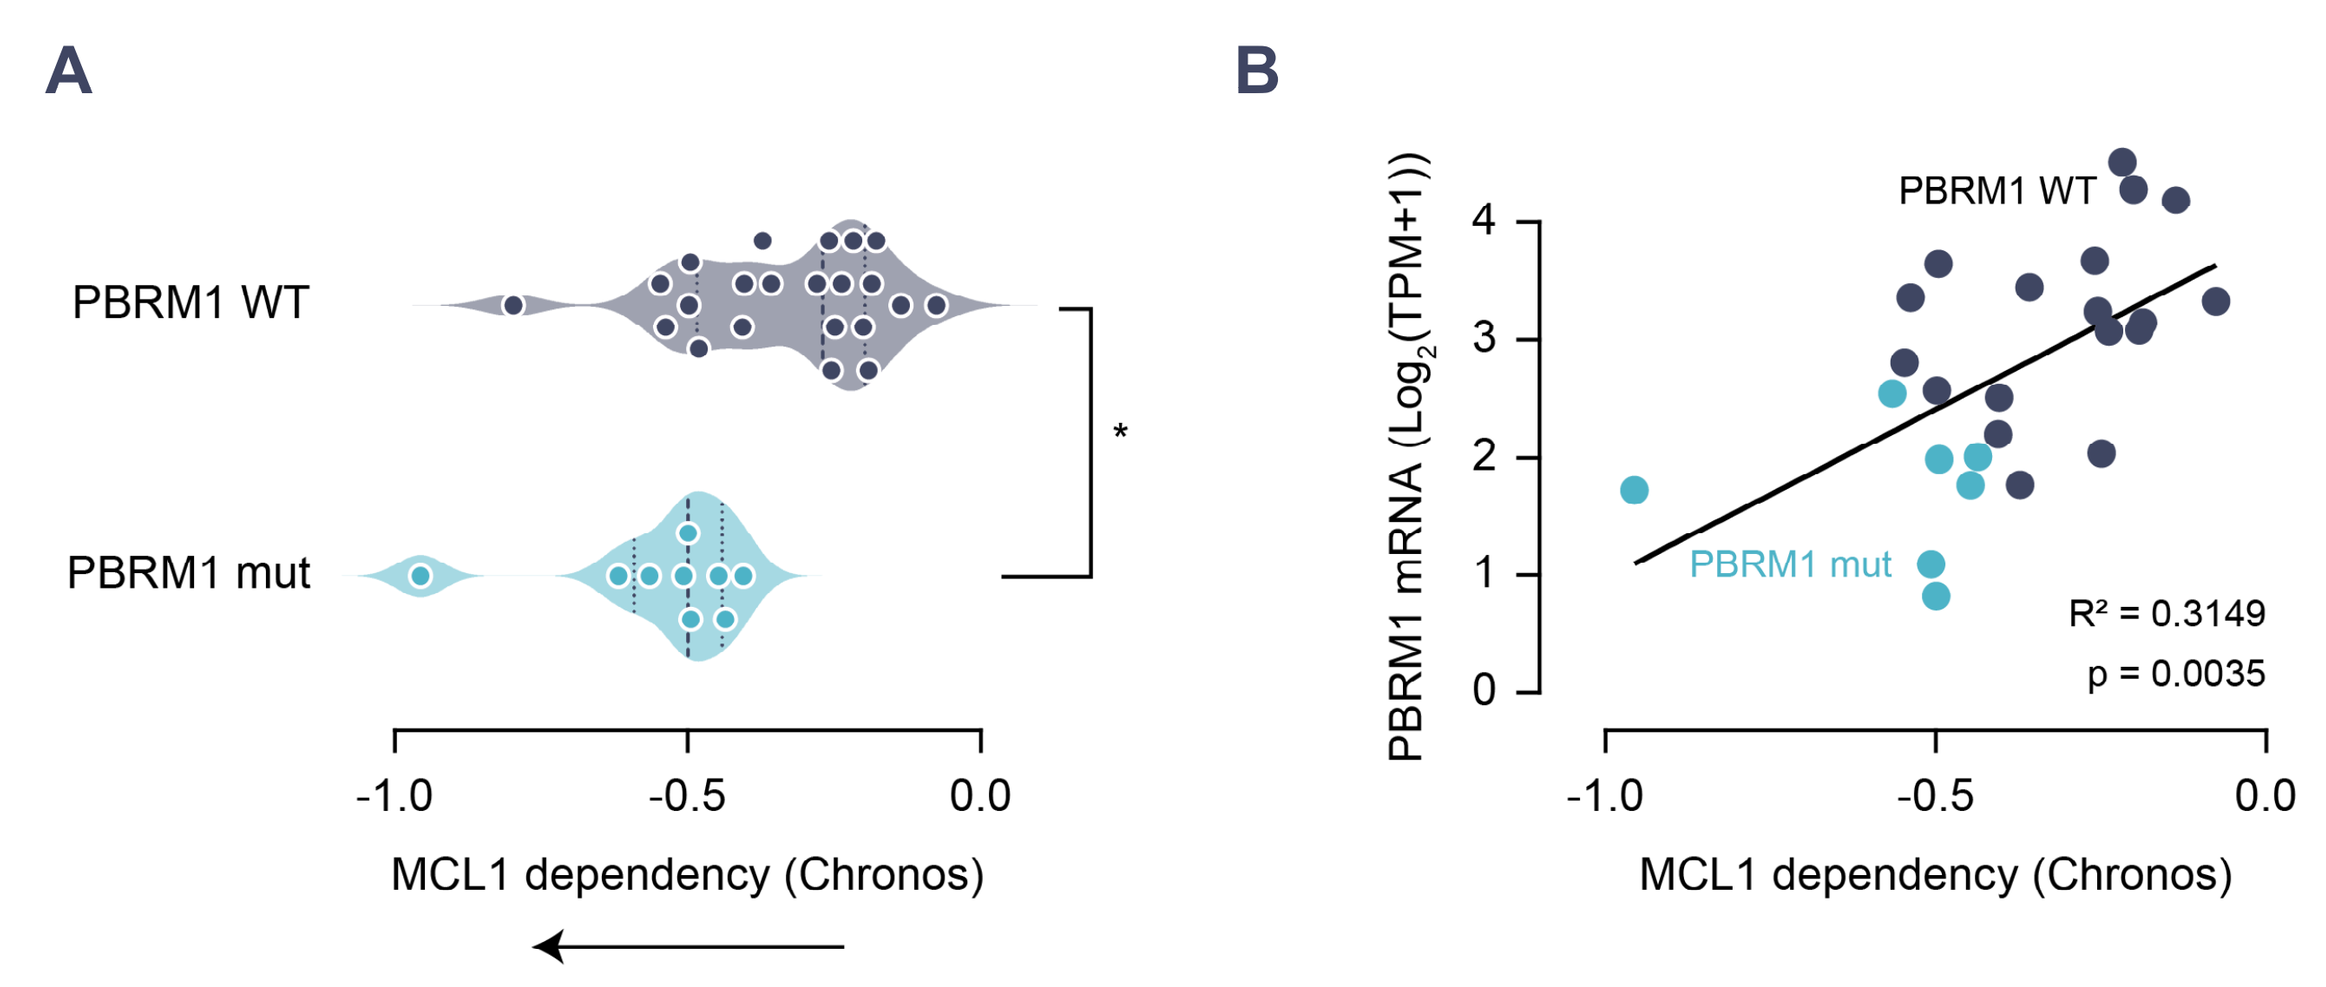
A
PBRM1 WT
PBRM1 mut
*
-1.0
-0.5
0.0
MCL1 dependency (Chronos)
B
PBRM1 mRNA (Log2(TPM+1))
PBRM1 WT
PBRM1 mut
R² = 0.3149
p = 0.0035
MCL1 dependency (Chronos)

(A) MCL-1 gene dependency analysis in ccRCC cell lines with damaging PBRM1 mutations compared to WT. Dependency scores (Chronos, 22Q2) were retrieved from DepMap (https://depmap.org) **(B)**. PBRM1 mRNA levels in ccRCC cell lines inversely correlate with MCL1 dependency. *P<0.05, versus WT by t-test.

Figure 2. ccRCC cell lines with PBRM1 mutations are sensitive to PRT1419 *in vitro*

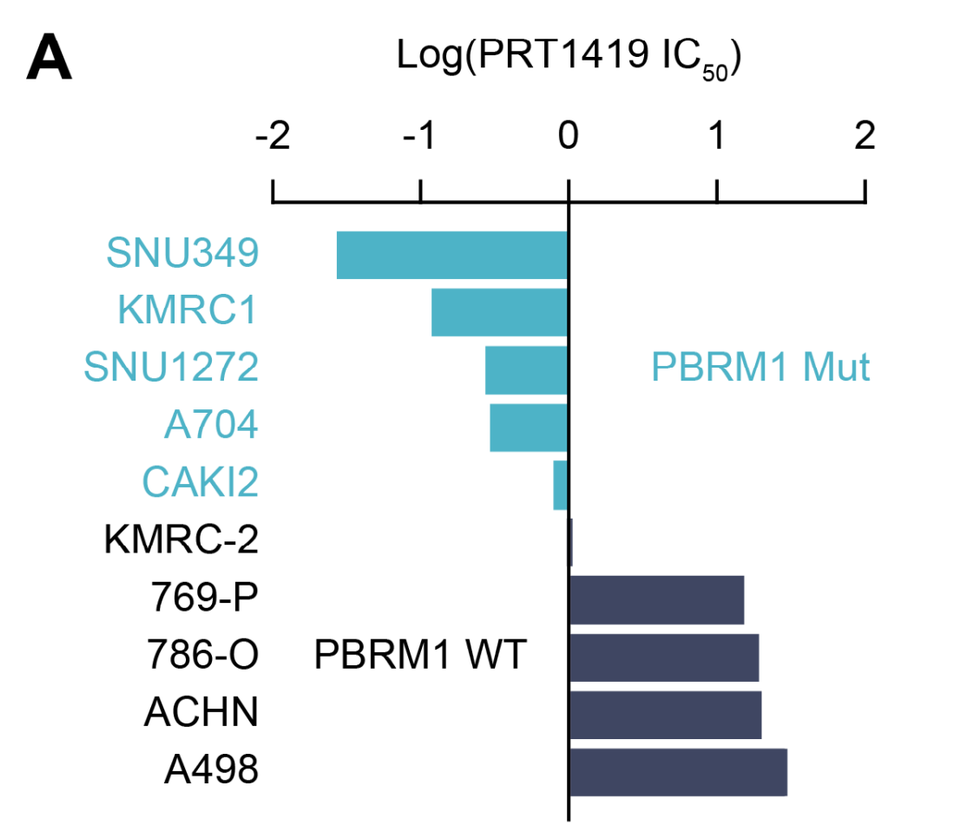
A
Log(PRT1419 IC50)
-2
-1
0
1
2
SNU349
KMRC1
SNU1272
A704
CAKI2
PBRM1 Mut
KMRC-2
769-P
786-O
ACHN
A498
PBRM1 WT

(A) Waterfall plot showing half maximal inhibitory concentration (IC50) for PRT1419 in the ccRCC panel. **(B)** Cell Titer-Glo assay assessing inhibition of 3D spheroid growth in PBRM1 mutant and WT ccRCC cell lines following treatment with MCL-1 inhibitor PRT1419 (72h). Values shown are cell viability calculated as percentage of vehicle (DMSO) control. **(C)** PRT1419 treatment induces concentration-dependent increase in cleaved Caspase 3/7 activity in PBRM1-mut lines but not WT lines. **(D, E)** PBRM1 depletion via RNAi in 769-P (PBRM1 WT ccRCC) induces sensitivity to PRT1419.

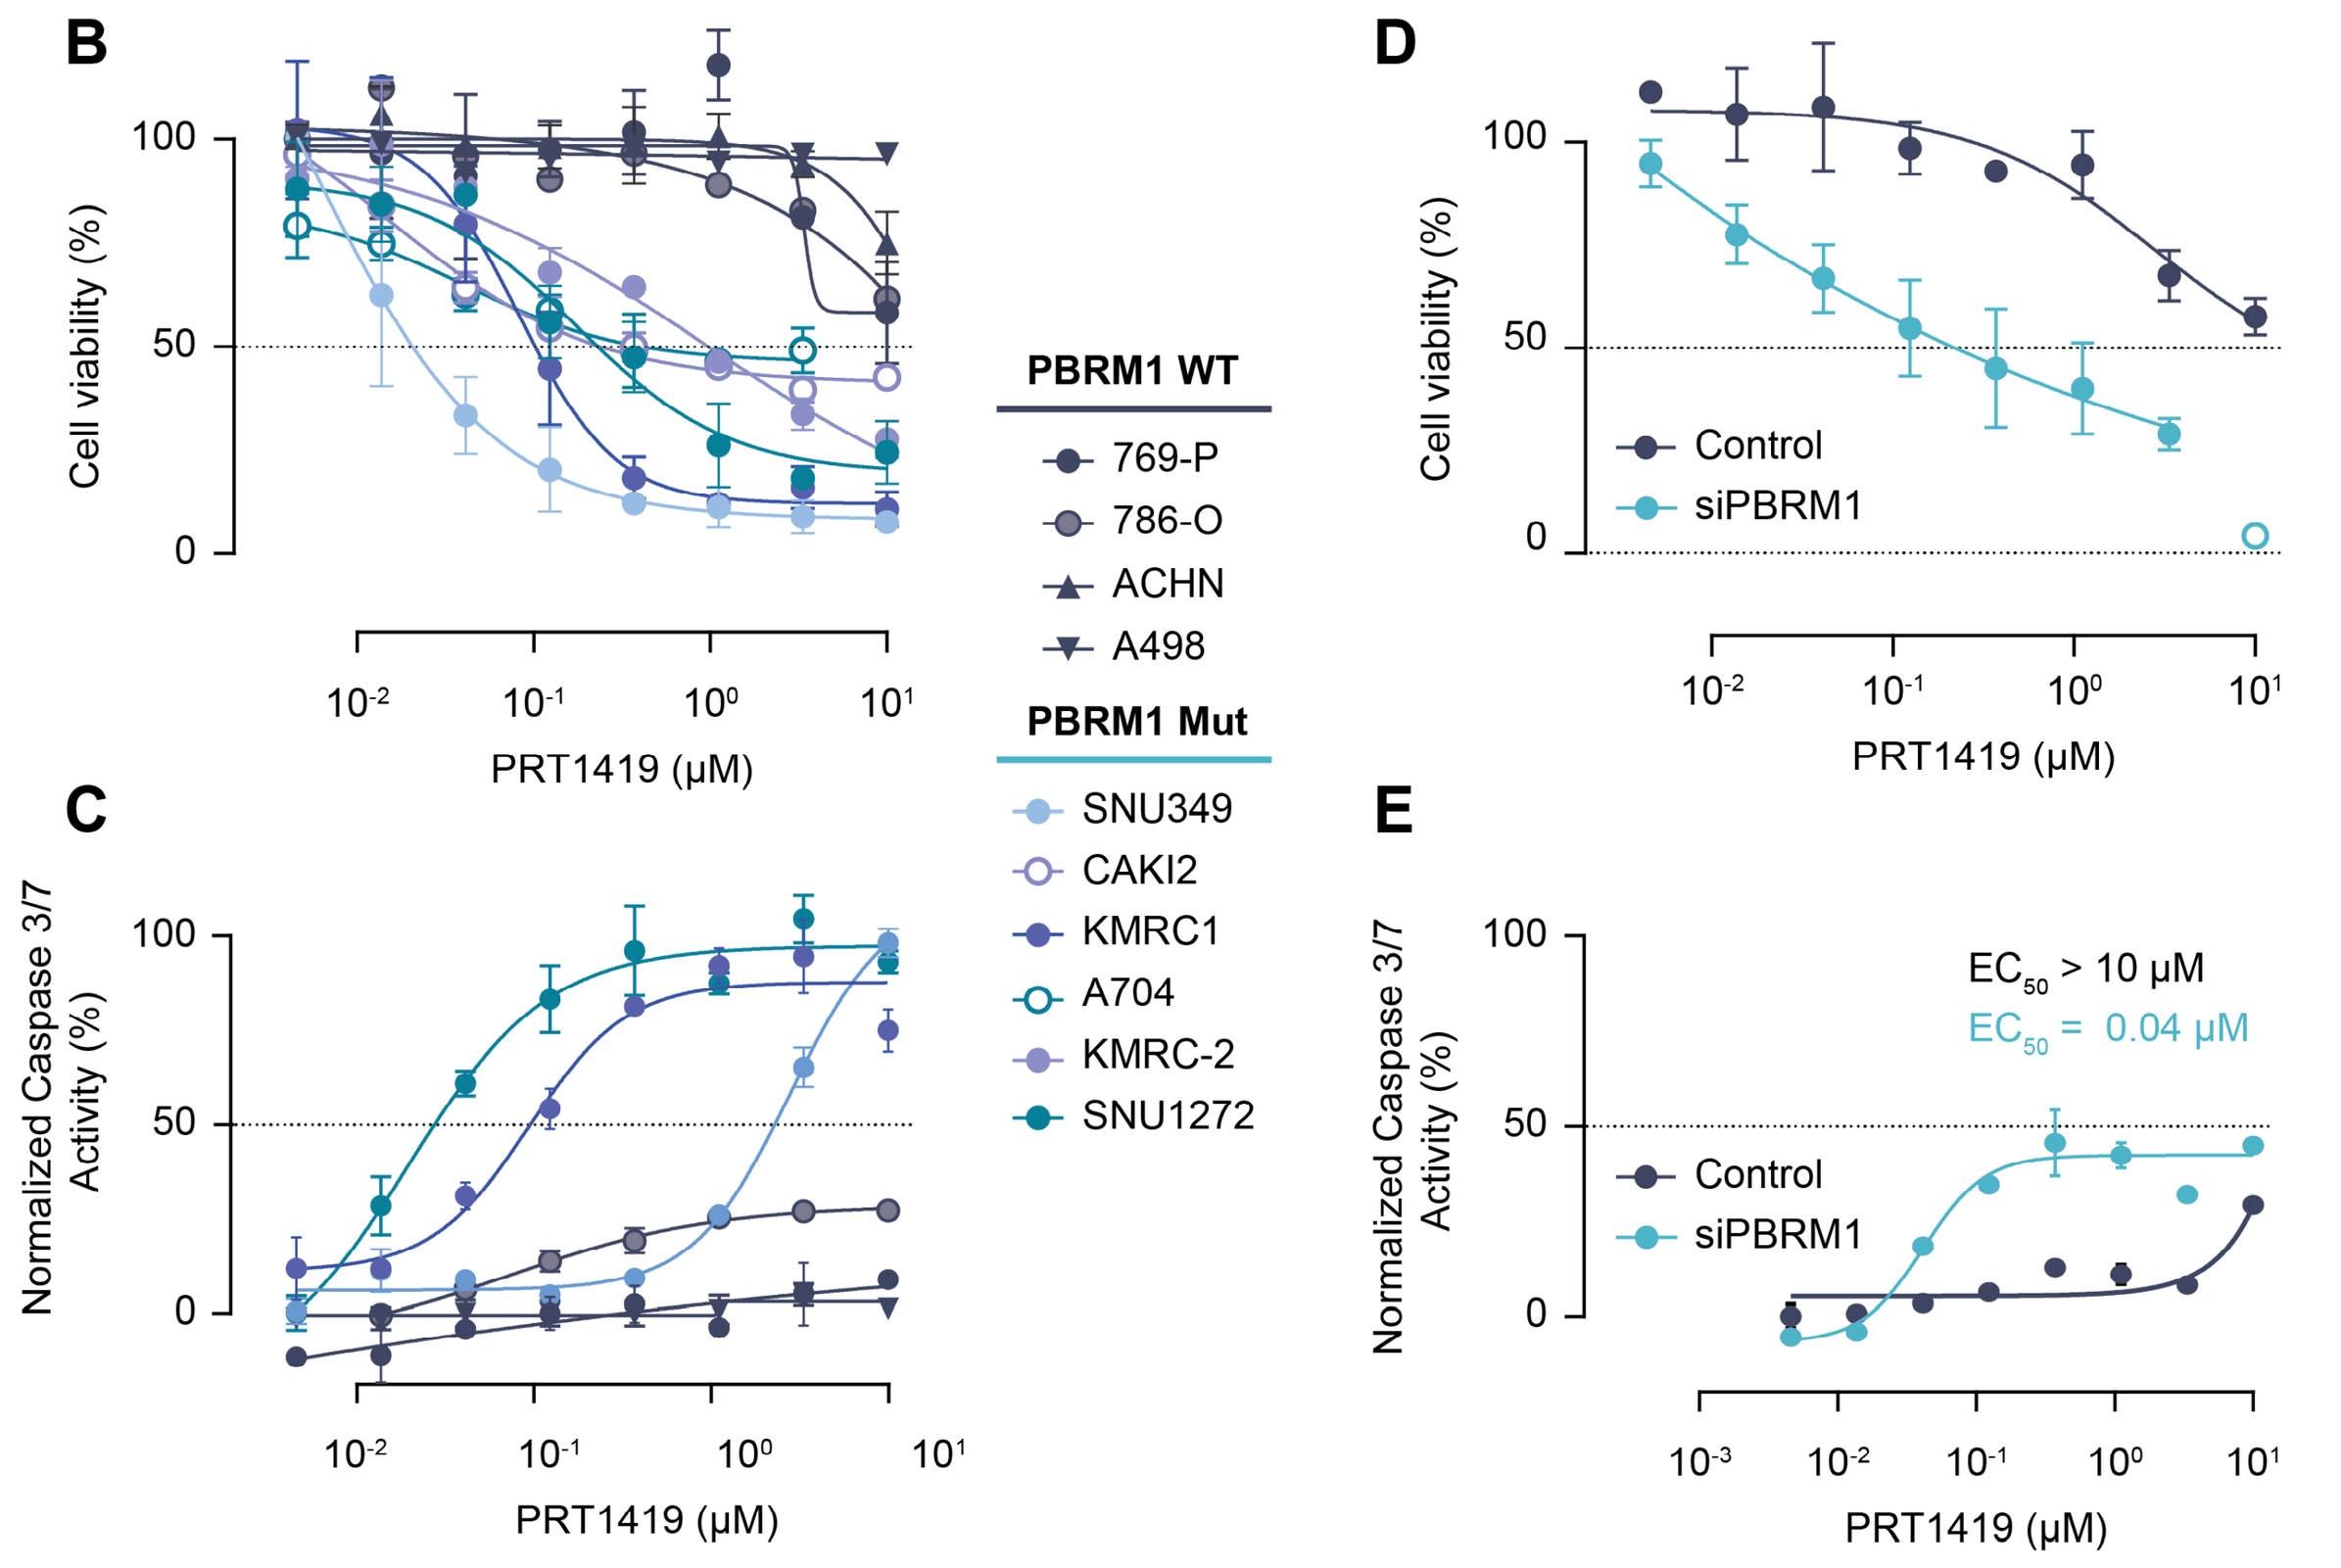
B
Cell viability (%)
PRT1419 (μM)
PBRM1 WT
769-P
786-O
ACHN
A498
PBRM1 Mut
SNU349
CAKI2
KMRC1
A704
KMRC-2
SNU1272
C
Normalized Caspase 3/7 Activity (%)
PRT1419 (μM)
D
Cell viability (%)
Control
siPBRM1
PRT1419 (μM)
E
Normalized Caspase 3/7 Activity (%)
EC50 > 10 μM
EC50 = 0.04 μM
Control
siPBRM1
PRT1419 (μM)

Figure 3. PBRM1 loss in ccRCC is associated with pro-apoptotic signaling, priming cells for cell death following treatment with PRT1419

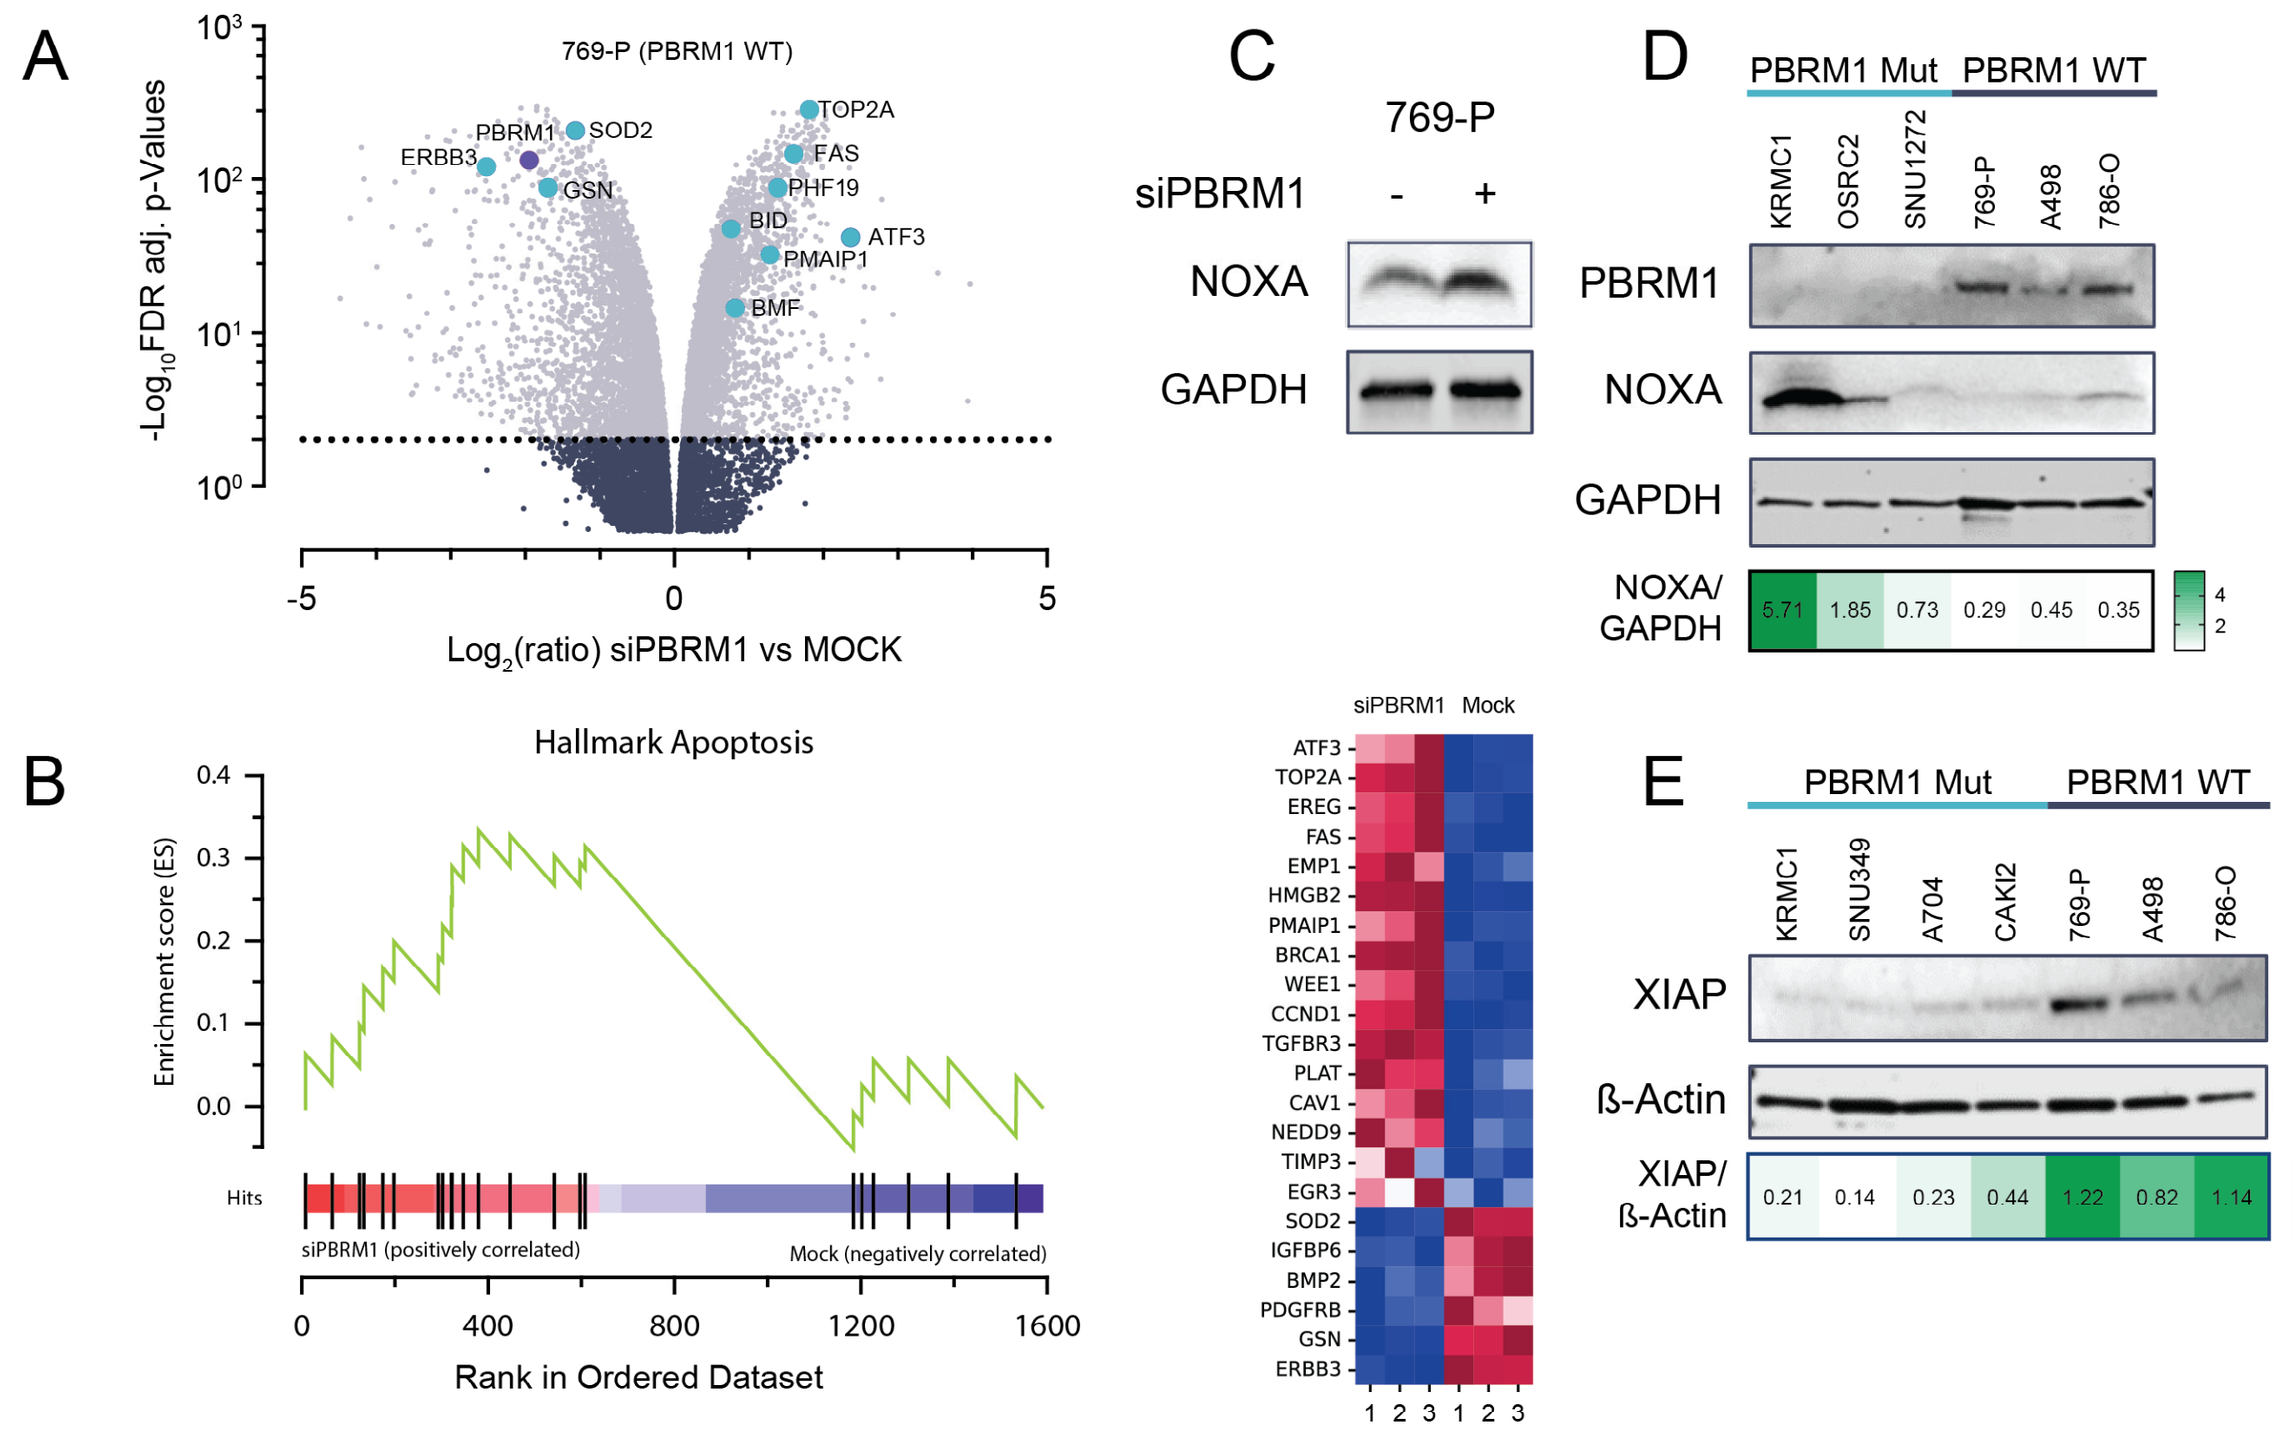
A
769-P (PBRM1 WT)
-Log10FDR adj. p-Values
ERBB3
PBRM1
SOD2
GSN
TOP2A
FAS
PHF19
BID
ATF3
PMAIP1
BMF
Log2(ratio) siPBRM1 vs MOCK
B
Hallmark Apoptosis
Enrichment score (ES)
Hits
siPBRM1 (positively correlated)
Mock (negatively correlated)
Rank in Ordered Dataset
siPBRM1
Mock
ATF3
TOP2A
EREG
FAS
EMP1
HMGB2
PMAIP1
BRCA1
WEE1
CCND1
TGFBR3
PLAT
CAV1
NEDD9
TIMP3
EGR3
SOD2
IGFBP6
BMP2
PDGFRB
GSN
ERBB3
C
769-P
siPBRM1
-
+
NOXA
GAPDH
D
PBRM1 Mut
PBRM1 WT
KRMC1
OSRC2
SNU1272
769-P
A498
786-O
PBRM1
NOXA
GAPDH
NOXA/ GAPDH
5.71
1.85
0.73
0.29
0.45
0.35
E
PBRM1 Mut
PBRM1 WT
KRMC1
SNU349
A704
CAKI2
769-P
A498
786-O
XIAP
ß-Actin
XIAP/ ß-Actin
0.21
0.14
0.23
0.44
1.22
0.82
1.14

(A) Volcano plots of global RNA expression data in 769-P (PBRM1 WT ccRCC) cells following RNAi-mediated PBRM1 depletion and 24h spheroid culture; genes mediating apoptosis are highlighted in blue. **(B)** Unbiased Gene Set Enrichment Analysis (GSEA) analysis (MSigDB Hallmark geneset)[6] demonstrates that PBRM1 depletion increases expression of a pro-apoptosis gene signature in ccRCC. **(C)** Western blot showing increased expression of pro-apoptotic MCL1 inhibitor, NOXA (*PMAIP1*), following PBRM1 depletion. **(D, E)** Western blot showing increased endogenous expression of NOXA and reduced expression of XIAP, a caspase inhibitor, in PBRM1 mutant ccRCC cell lines cultured as spheroids, suggesting apoptosis "priming."

Figure 4. PRT1419 synergizes with Standard of Care (SoC) agents in PBRM1-mutant ccRCC

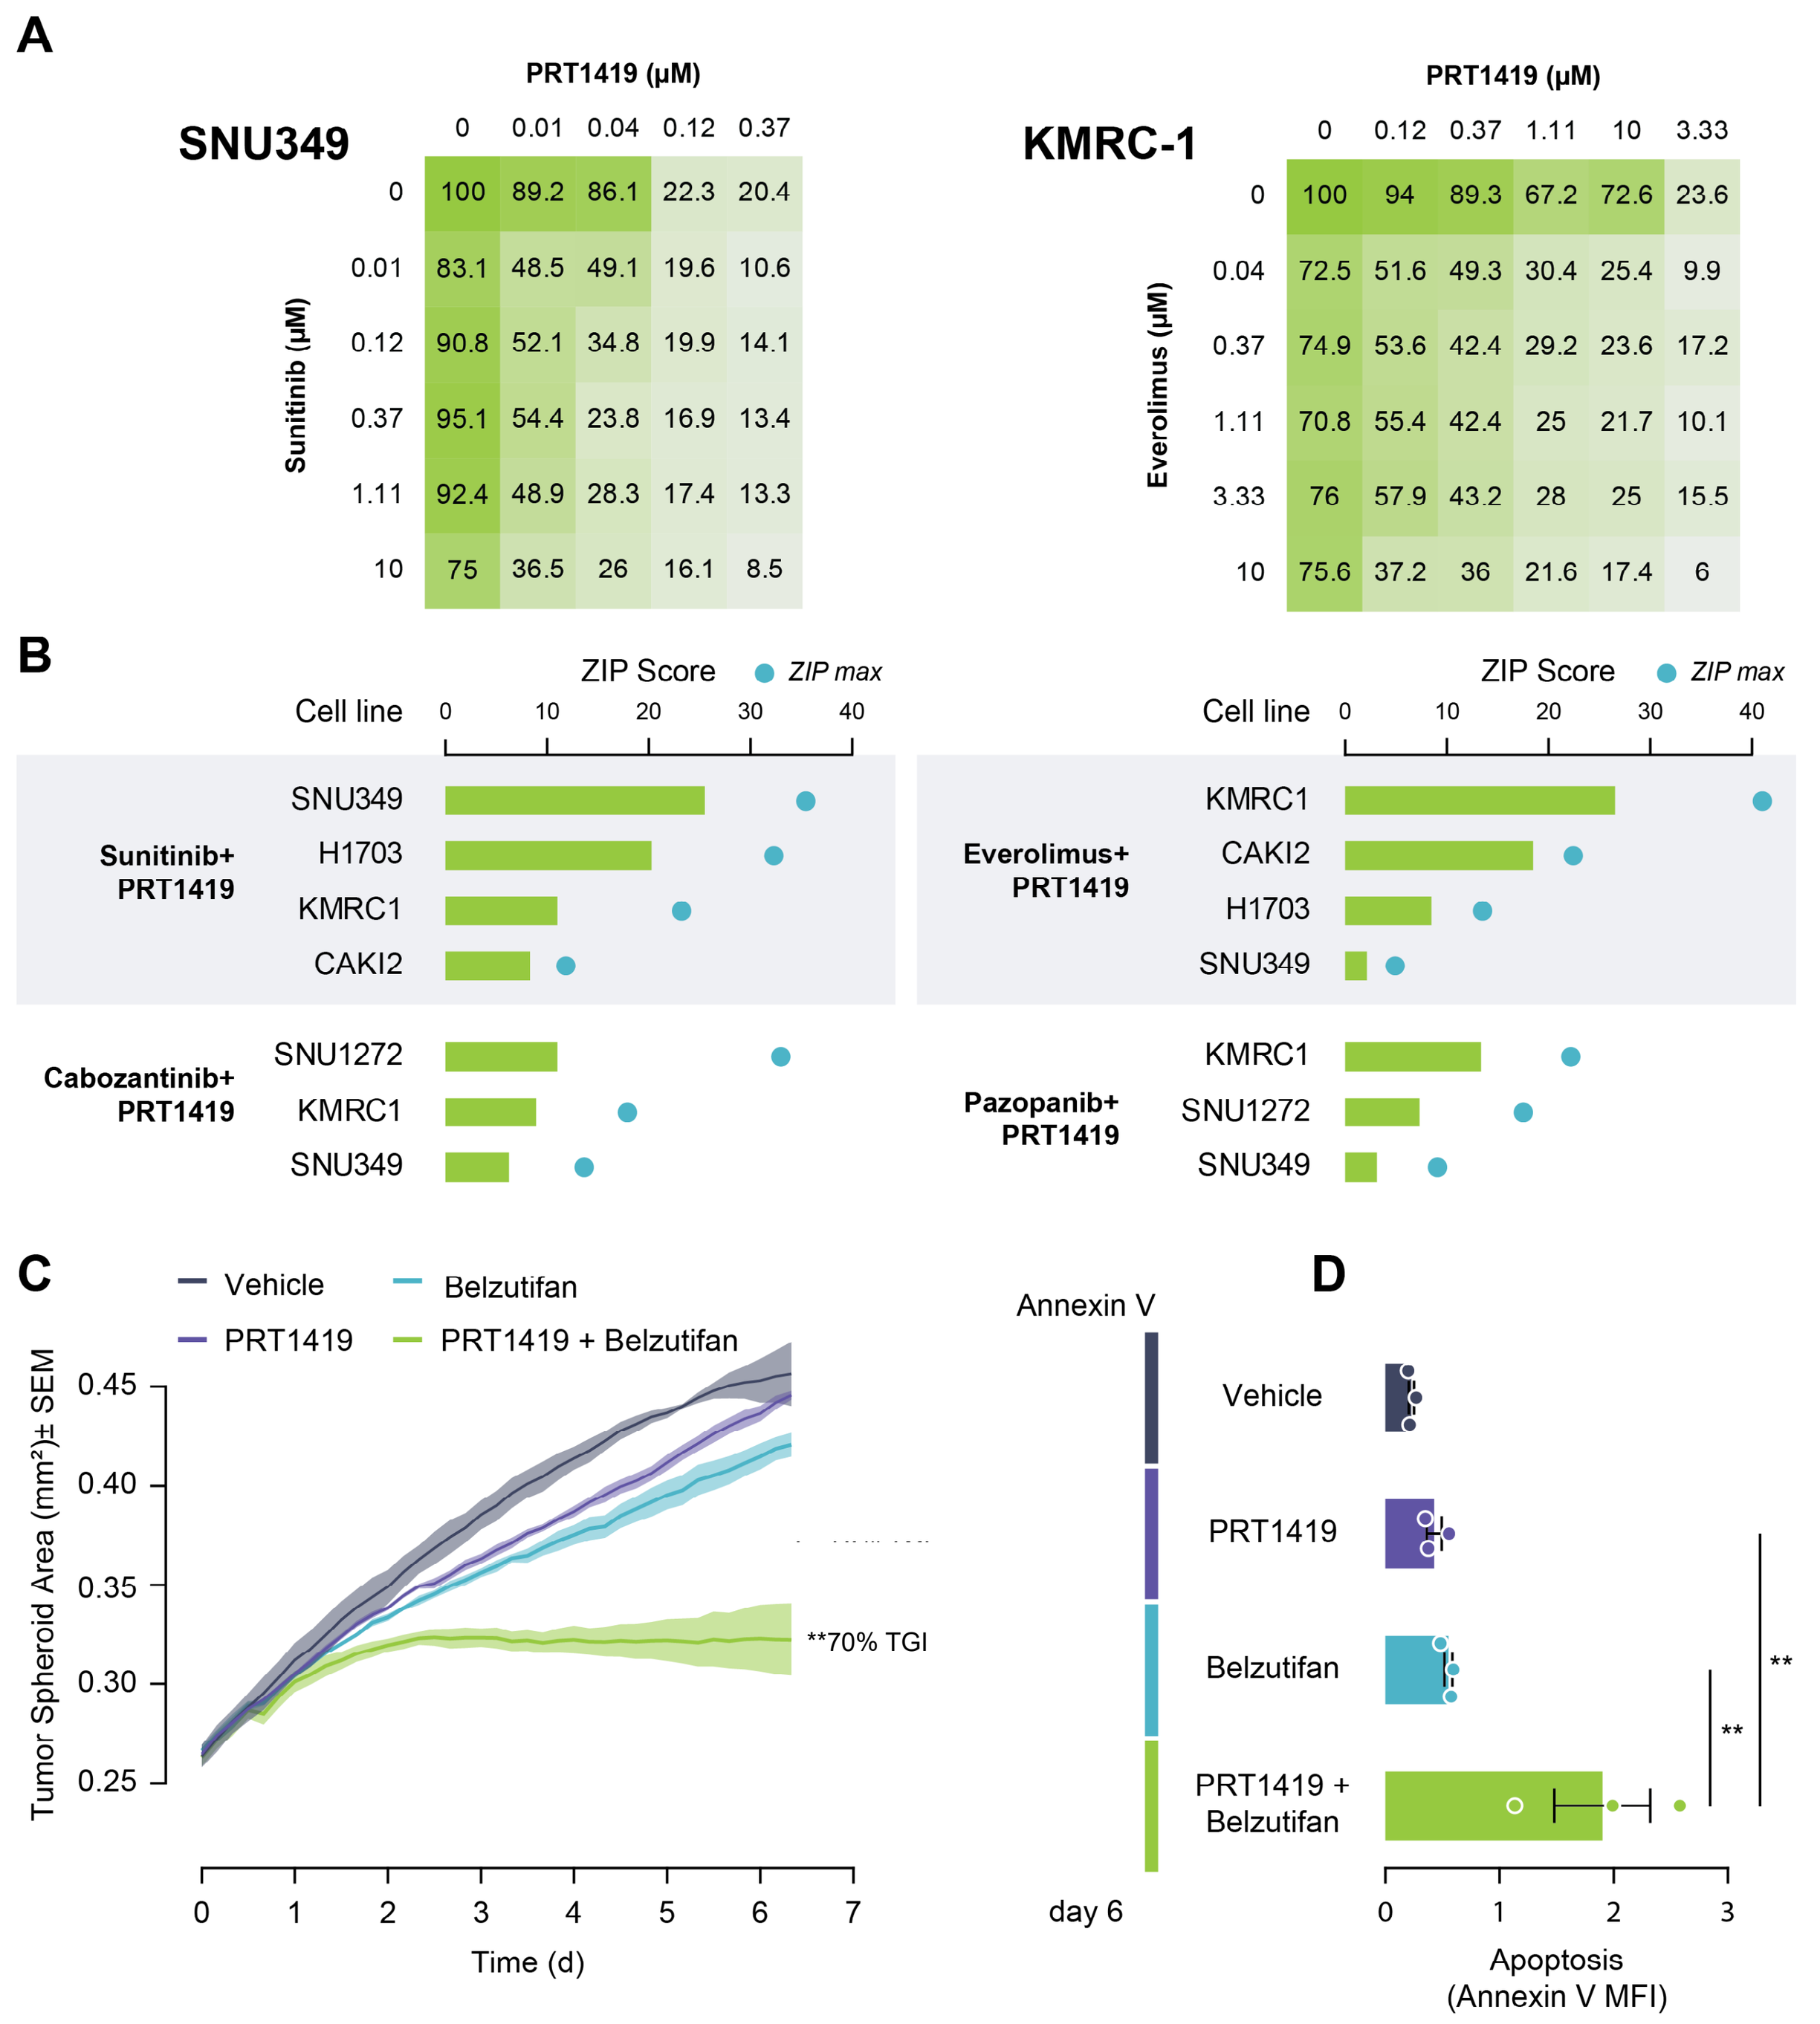
A
SNU349
PRT1419 (μM)
Sunitinib (μM)
0 0.01 0.04 0.12 0.37
0: 100 89.2 86.1 22.3 20.4
0.01: 83.1 48.5 49.1 19.6 10.6
0.12: 90.8 52.1 34.8 19.9 14.1
0.37: 95.1 54.4 23.8 16.9 13.4
1.11: 92.4 48.9 28.3 17.4 13.3
10: 75 36.5 26 16.1 8.5
KMRC-1
PRT1419 (μM)
Everolimus (μM)
0 0.12 0.37 1.11 10 3.33
0: 100 94 89.3 67.2 72.6 23.6
0.04: 72.5 51.6 49.3 30.4 25.4 9.9
0.37: 74.9 53.6 42.4 29.2 23.6 17.2
1.11: 70.8 55.4 42.4 25 21.7 10.1
3.33: 76 57.9 43.2 28 25 15.5
10: 75.6 37.2 36 21.6 17.4 6
B
ZIP Score
ZIP max
Cell line
Sunitinib+ PRT1419
SNU349
H1703
KMRC1
CAKI2
Cabozantinib+ PRT1419
SNU1272
KMRC1
SNU349
Everolimus+ PRT1419
KMRC1
CAKI2
H1703
SNU349
Pazopanib+ PRT1419
KMRC1
SNU1272
SNU349
C
Vehicle
PRT1419
Belzutifan
PRT1419 + Belzutifan
Tumor Spheroid Area (mm²)± SEM
**70% TGI
Time (d)
D
Annexin V
Vehicle
PRT1419
Belzutifan
PRT1419 + Belzutifan
day 6
Apoptosis (Annexin V MFI)

A) CellTiter-Glo assay demonstrating synergistic inhibition of spheroid growth in PBRM1-mutant cell lines following treatment with PRT1419 and SoC agents: Sunitinib, Everolimus, Cabozantinib and Pazopanib. Values shown are cell viability calculated as percentage of vehicle control. **(B)** ZIP synergy scores from synergy proliferation assays were computed using SynergyFinder 2.0 (https://synergyfinder.fimm.fi/). **(C, D)** Incucyte-based measurement of spheroid growth and apoptosis (Annexin V) in KMRC2 (VHL protein loss, PBRM1 mutant) following treatment with PRT1419 (0.5 μM) and/or Belzutifan (HIF2α inhibitor, 1 μM) for 6 days. Data represented as mean + SEM. N=3, ** P<0.01 (A) one-way ANOVA

Figure 5. Combination of PRT1419 and S.o.C agents leads to improved and durable response in vivo

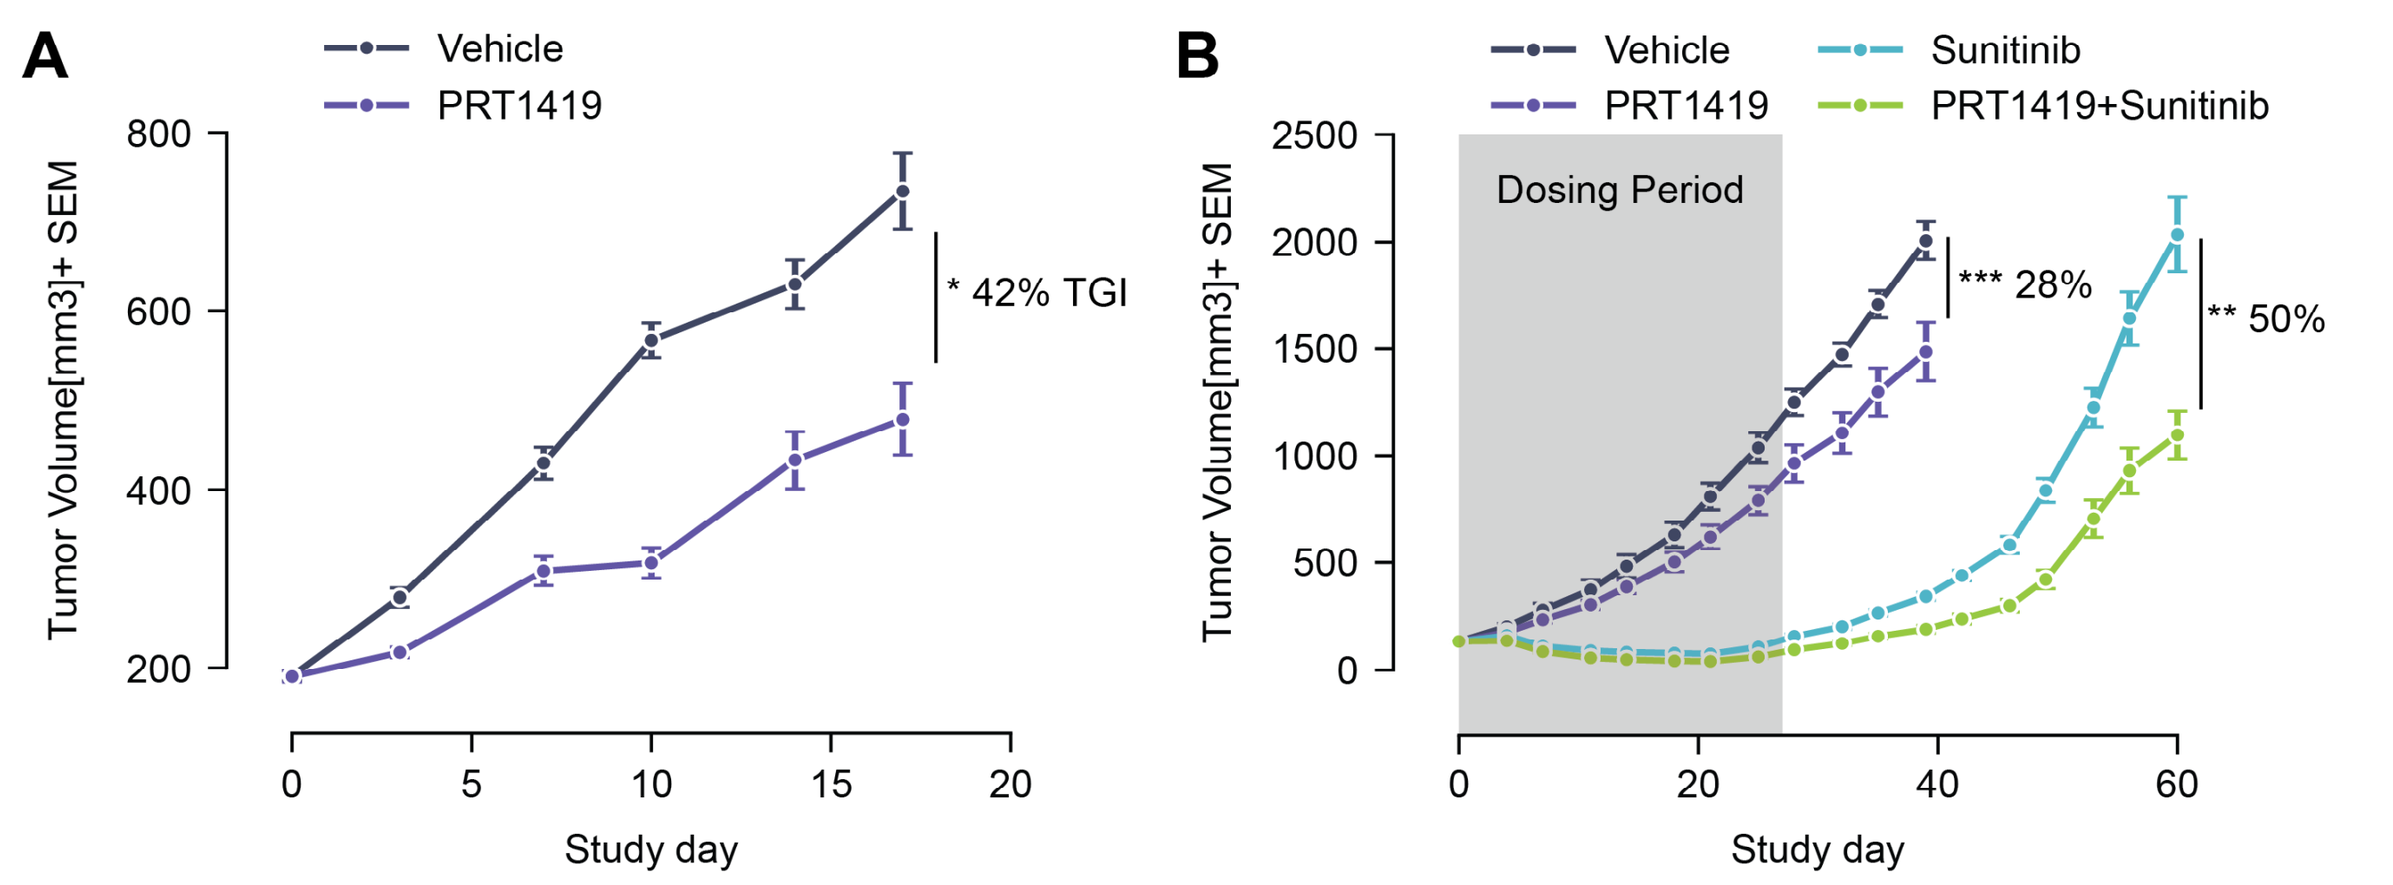
A
Vehicle
PRT1419
Tumor Volume[mm3]+ SEM
* 42% TGI
Study day
B
Vehicle
PRT1419
Sunitinib
PRT1419+Sunitinib
Dosing Period
Tumor Volume[mm3]+ SEM
*** 28%
** 50%
Study day

Intravenously administered PRT1419 is efficacious in monotherapy and in combination with Sunitinib, administered orally, to potently repress tumor growth in CDX models of PBRM1-loss ccRCC and NSCLC. **(A)** Female BALB/c nude mice were injected subcutaneously with OSRC-2 (ccRCC, PBRM1 mutant) in the right front flank region (1 x 10[6] cells) **(B)** Female NOD/SCID mice were injected subcutaneously in the right front flank region with PBRM1-protein loss NSCLC cell line NCI-H1703 (1 x 10[7] cells). Animals were dosed with PRT1419 once a week and/or Sunitinib(10 mg/kg) daily. Data represented as mean + SEM. N=8, * P<0.05, **P<0.001, ***P<0.001 by (A) Mann-Whitney U test or (B) unpaired t test

Figure 6. PRT2527, a potent and selective CDK9 inhibitor, similarly demonstrates antitumor activity in PBRM1-loss ccRCC

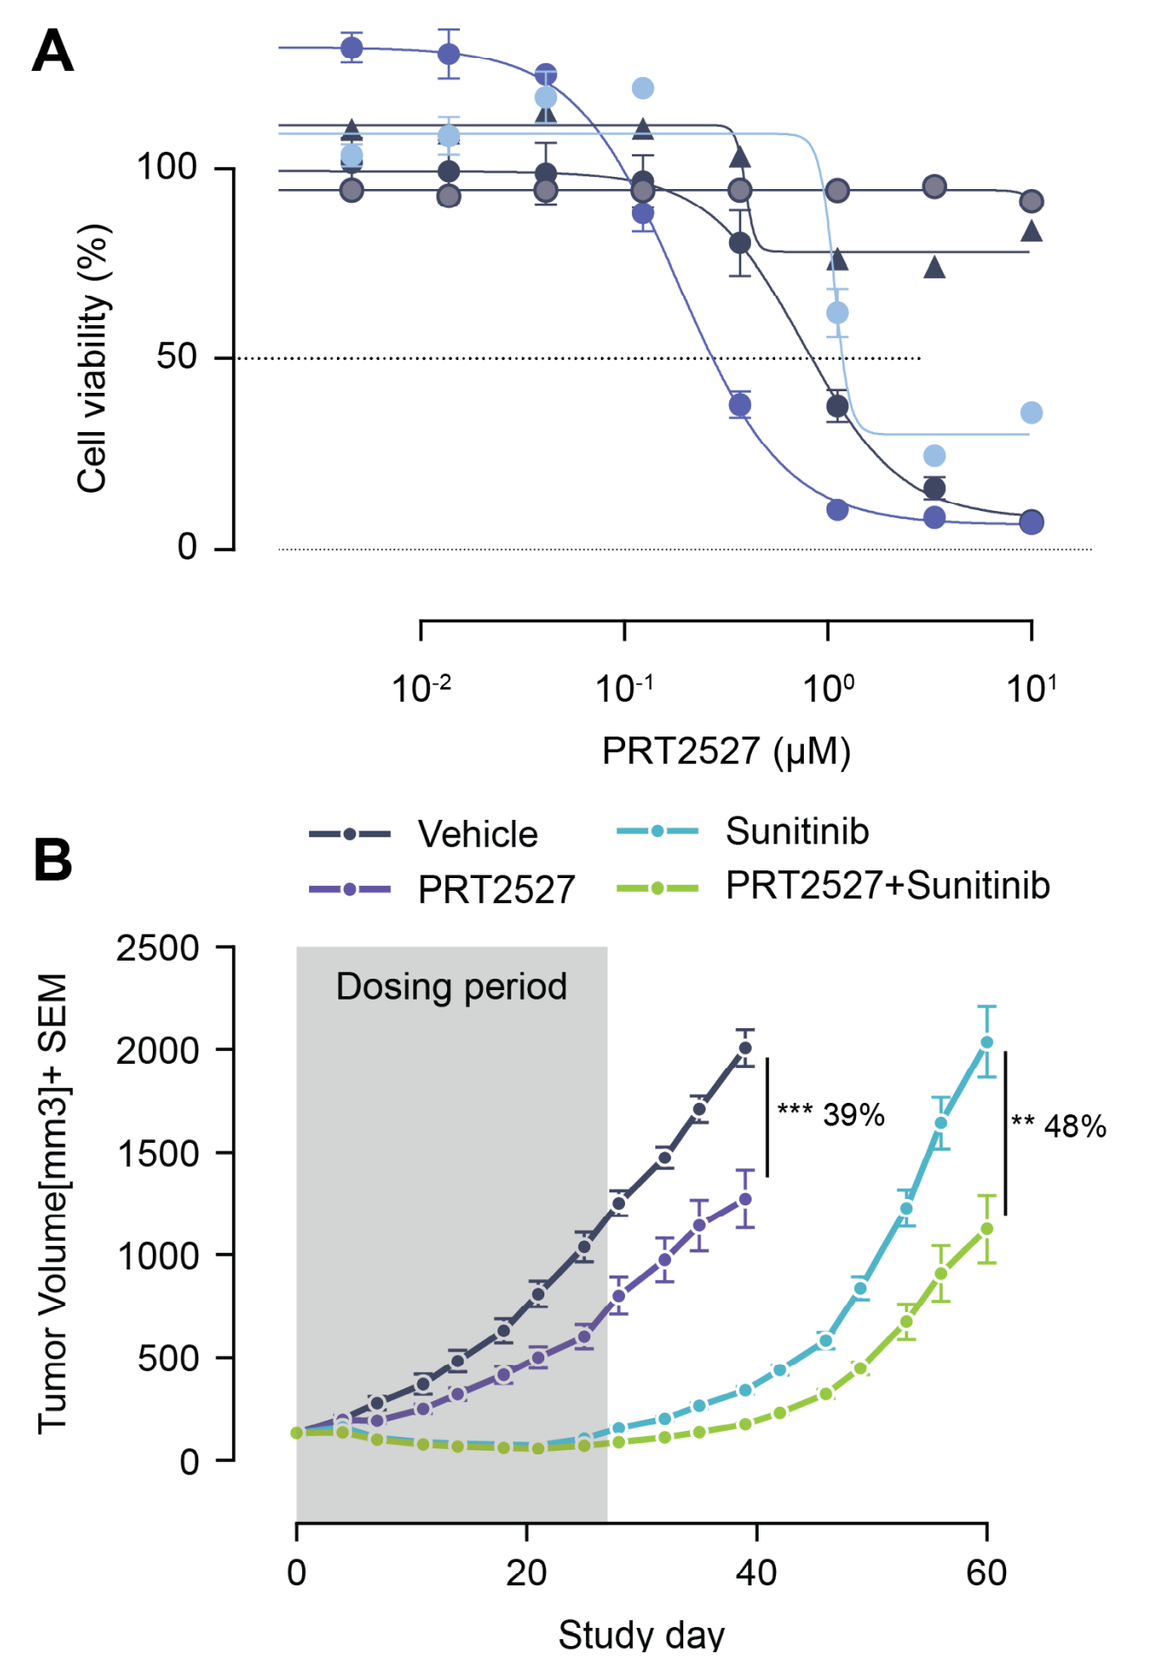
A
Cell viability (%)
PRT2527 (μM)
PBRM1 WT
769-P MYCamp
786-O
ACHN
PBRM1 Mut
SNU349
KMRC1
B
Vehicle
PRT2527
Sunitinib
PRT2527+Sunitinib
Dosing period
Tumor Volume[mm3]+ SEM
*** 39%
** 48%
Study day

Figure 6. (A) CellTiter-Glo assay assessing inhibition of spheroid growth in PBRM1 mutant and WT ccRCC cell lines following brief treatment with CDK9 inhibitor PRT2527. Cells grown as spheroids were treated for 4h, then cultured in drug-free media for 48h **(B)** Intravenously administered PRT2527 combines with Sunitinib, dosed orally, to repress tumor growth in a PBRM1-protein loss NSCLC (NCI-H1703) cell line-derived xenograft model. Female NOD/SCID mice were injected subcutaneously in the right front flank region with NCI-H1703 cells(1 x 10[7] cells). Animals were dosed with PRT2527 once a week (twice in one day) and/or Sunitinib (10 mg/kg) daily. Data represented as mean + SEM. N=8, **P<0.001 by unpaired t test

Conclusions

- PBRM1 loss is associated with sensitivity to MCL1 inhibition in preclinical models of ccRCC
- PBRM1 loss is associated with pro-apoptotic signaling, priming cells for apoptosis following MCL1 inhibition.
- PRT1419 synergizes with SoC (multitargeted TKIs, mTOR inhibitors and HIF2α inhibitor) in inhibiting tumor growth *in vitro* and *in vivo*.
- Indirect inhibitors of MCL1 expression, such as selective CDK9 inhibitor PRT2527, are similarly efficacious in PBRM1-deficient ccRCC.
- PRT1419 and PRT2527 are currently being evaluated in Phase I clinical trials in patients with relapsed/refractory hematologic and solid malignancies.[2,7]

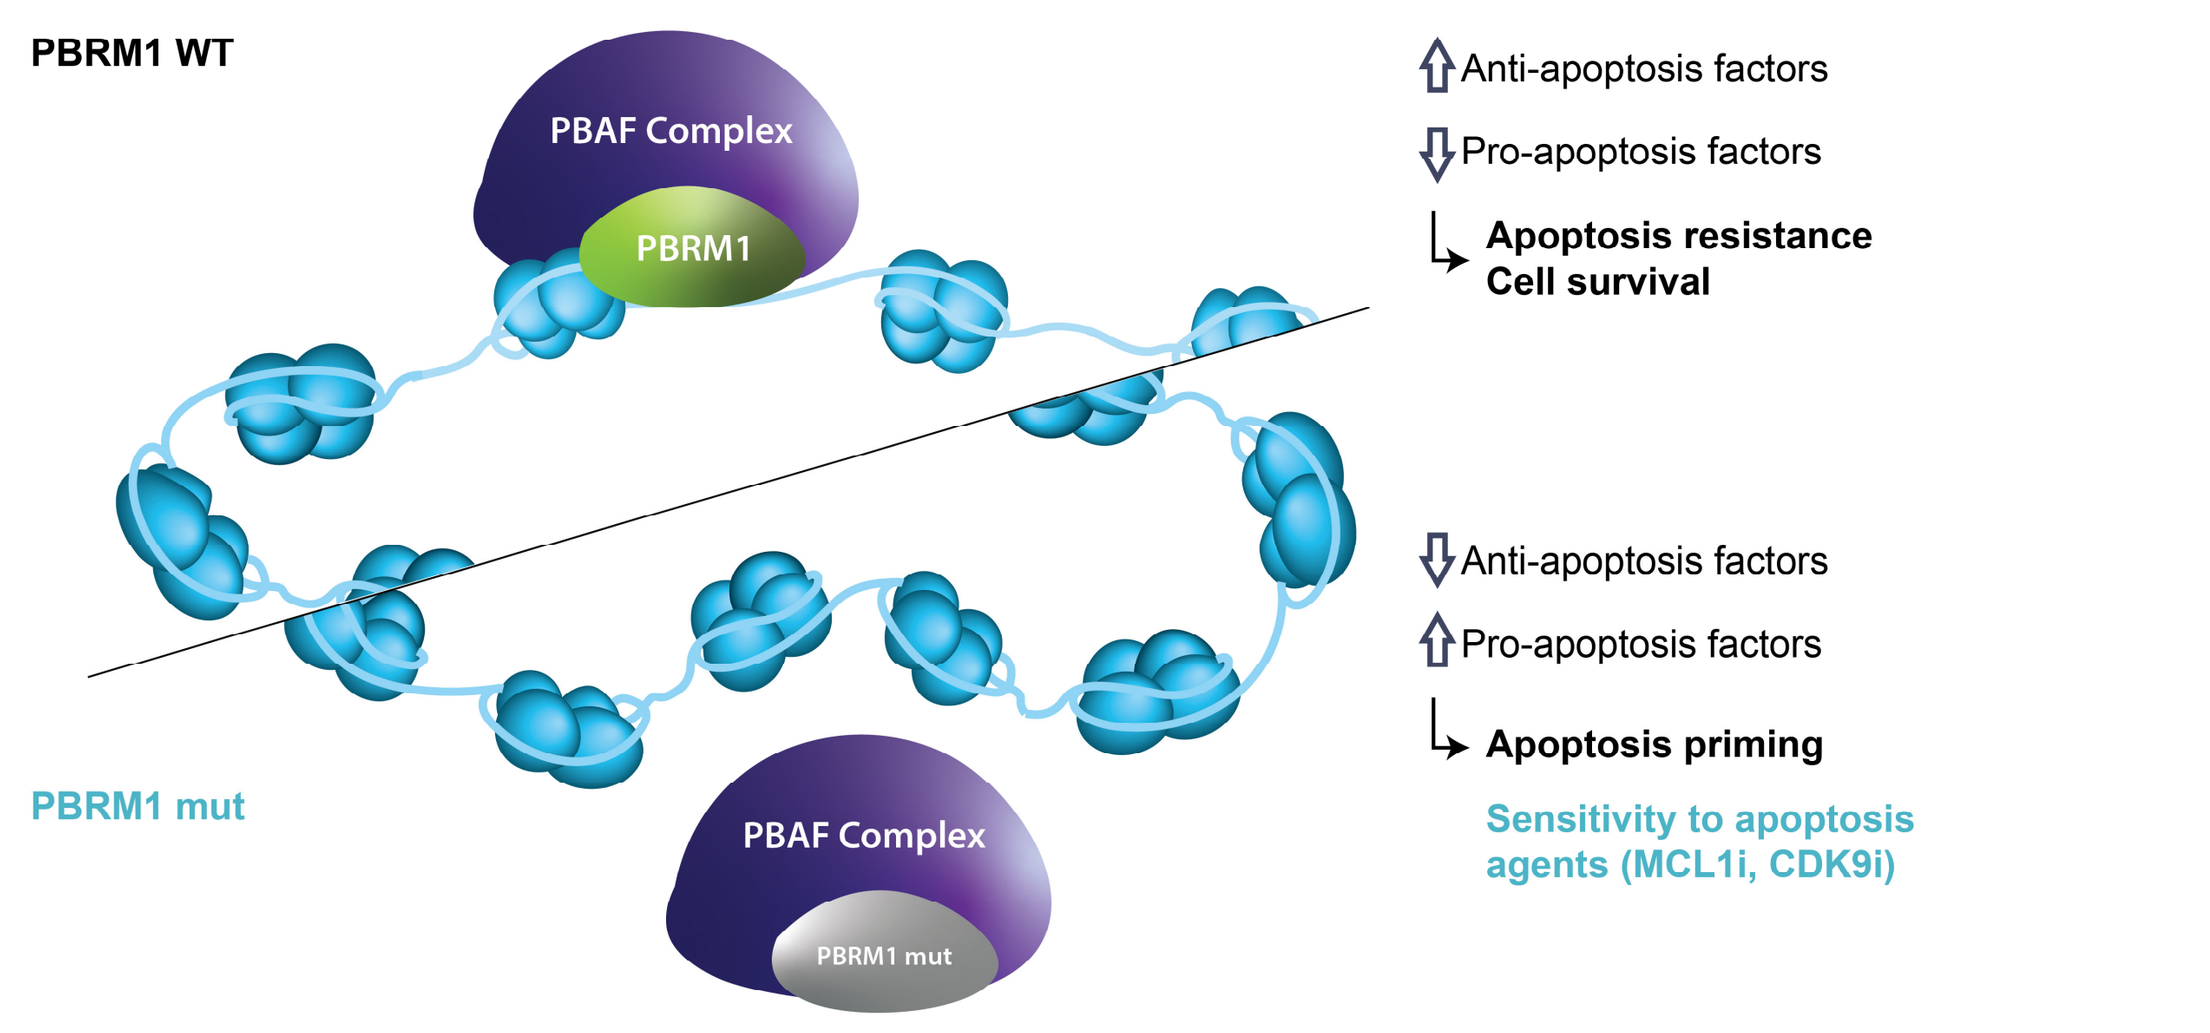
PBRM1 WT
PBAF Complex
PBRM1
Anti-apoptosis factors
Pro-apoptosis factors
Apoptosis resistance
Cell survival
Anti-apoptosis factors
Pro-apoptosis factors
Apoptosis priming
PBRM1 mut
PBAF Complex
PBRM1 mut
Sensitivity to apoptosis agents (MCL1i, CDK9i)

References

1. Campbell K.J, et al, 2018. *Open biology*, *8*(5), p.180002.
2. NCT05107856, NCT04837677 (ClinicalTrials.gov).
3. Fultang, N., et al, 2022. *Cancer Res*, 82(12_Supplement), pp.420-420.
4. Pawłowski R., et al, 2013. *Int. J. Cancer*, 132(2), pp.E11-E17
5. National Comprehensive Cancer Network. Kidney Cancer (Version 4.2023). Accessed: February 9th, 2023.
6. Liberzon A., et al, 2015. *Cell systems*, 1(6), pp.417-425.
7. NCT05665530, NCT05159518 (ClinicalTrials.gov).

Acknowledgements

This study was funded by Prelude Therapeutics, Inc. Editorial Support provided by Arne Fabritius, Endosymbiont GMBH, and funded by Prelude Therapeutics, Inc.

Disclosures

Authors are or were employees of Prelude Therapeutics, Inc at the time of research, and may own equity in the Company.